Rejuvenation: Innovative Technology to Improve T-cell Antitumor Properties Through Partial Reprogramming

Jessica Fioravanti[1], Yasuhiro Yamazaki[1], Takuya Maeda[1], Yin Huang[1], Naritaka Tamaoki[1], Kriti Bahl[1], Burak Kutlu[1], Shobha Potluri[1], Gary Lee[1], Nicholas P. Restifo[1], and Raul Vizcardo[1]

[1]Lyell Immunopharma, South San Francisco, CA 94080

Abstract No. A041

Methodology

Novel T-cell rejuvenation through partial reprogramming

Lyell's T-cell rejuvenation technology represents an innovative approach to cellular age reversal with exciting implications for the field of cancer immunotherapy.

Aging T cells are revitalized by the temporary expression of transcription factors typically associated with induced pluripotent stem cell (iPSC) reprogramming in a non-integrative fashion.

Our partial reprogramming is able to rejuvenate aging T cells without completely de-differentiating them into iPSCs, thus avoiding the complex and challenging process of redifferentiating iPSCs back into fully-restored T cells.

Lyell's data demonstrate that the resulting rejuvenated T (T_{RJ}) show:

- Younger epigenetic age (eAge)
- Enhanced cell proliferation and metabolism
- Improved antitumor potency
- Preservation of stemness markers and TCR repertoire.

Figure 1. Lyell's T cell rejuvenation technology to restore T-cell activity and enhance cancer immunotherapy

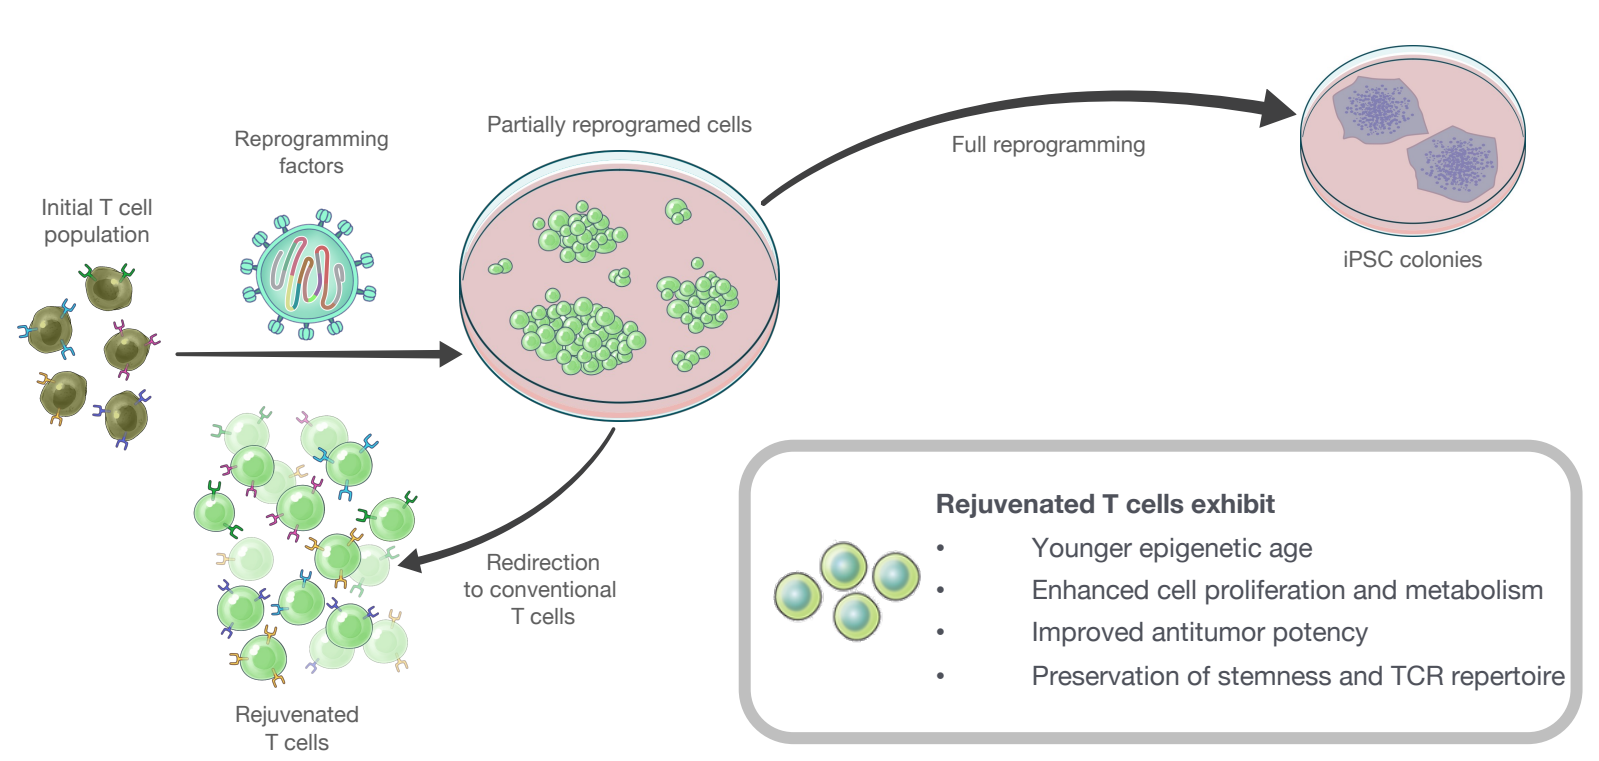
Initial T cell population
Reprogramming factors
Partially reprogramed cells
Full reprogramming
iPSC colonies
Redirection to conventional T cells
Rejuvenated T cells
Rejuvenated T cells exhibit
Younger epigenetic age
Enhanced cell proliferation and metabolism
Improved antitumor potency
Preservation of stemness and TCR repertoire

Results

Rejuvenated T cells are reinvigorated

Rejuvenated T cells (T_{RJ}) cells demonstrated a younger epigenetic age compared with non-rejuvenated control cells (T_{CT}) and also exhibited increased proliferation

Figure 2. Rejuvenated T cells from healthy donors show epigenetic youth and increased proliferation

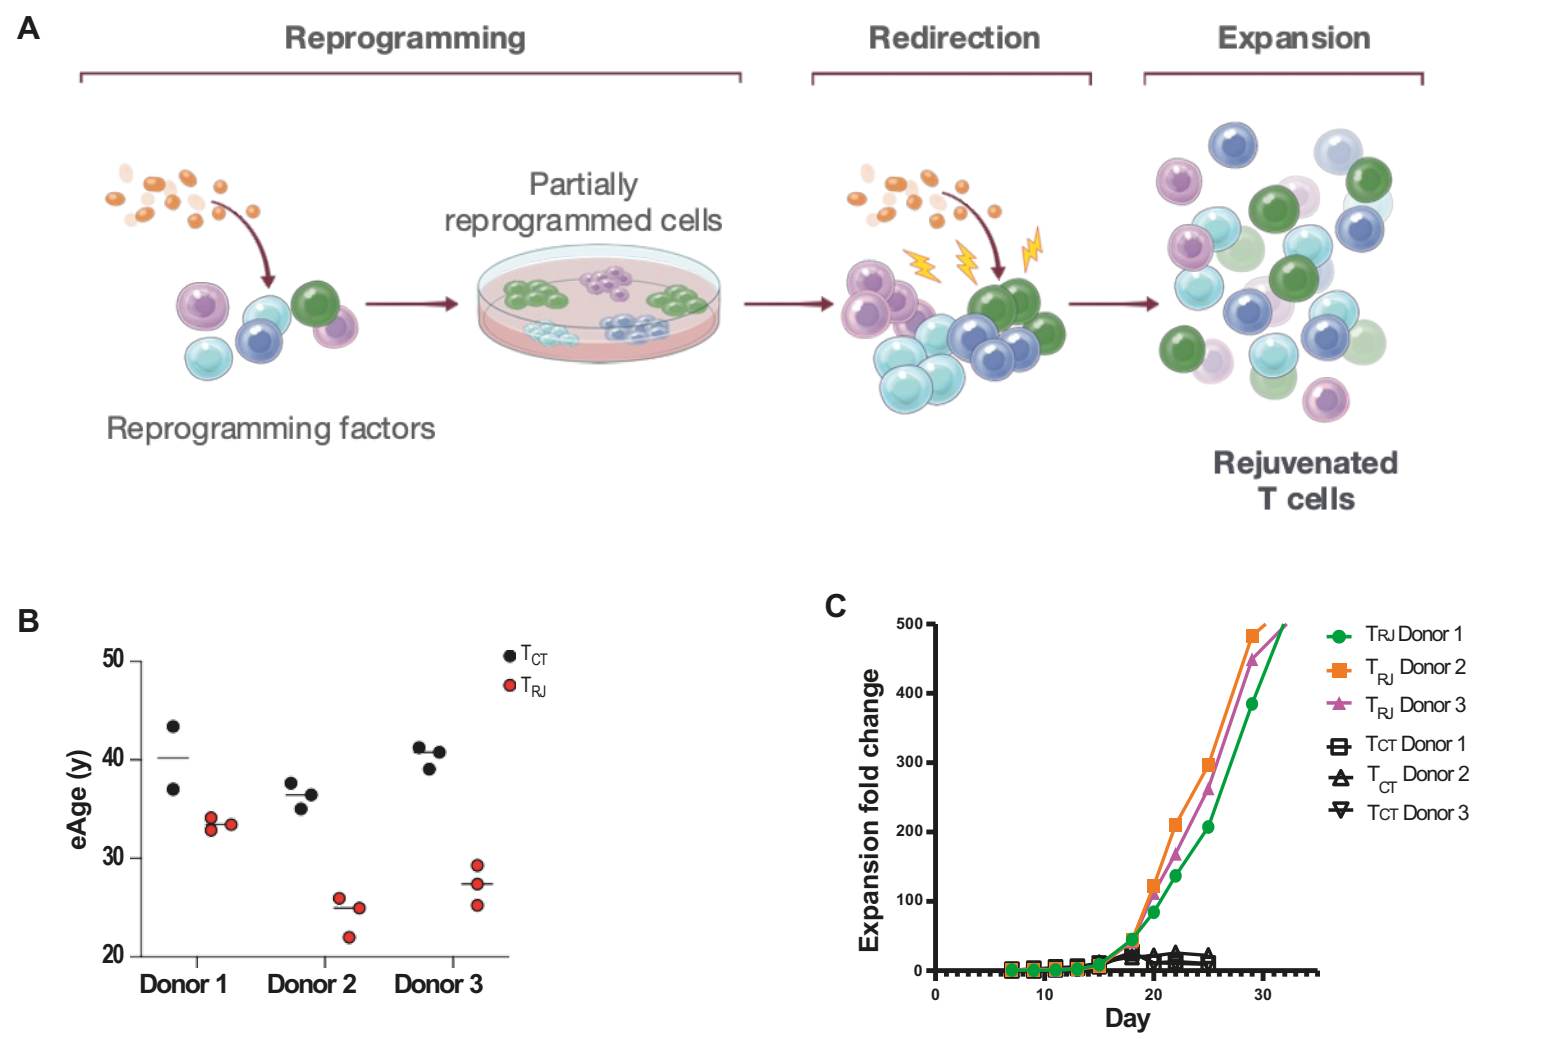
A
Reprogramming
Redirection
Expansion
Partially reprogrammed cells
Reprogramming factors
Rejuvenated T cells
B
eAge (y)
Donor 1
Donor 2
Donor 3
C
Expansion fold change
Day

(A) Schematic representation of T cell rejuvenation process. **(B and C)** Peripheral blood T cells from 3 healthy donors were subjected to partial reprogramming (Donor 1, 37 yo; Donor 2, 42 yo; Donor 3, 52 yo). Measurement of epigenetic age (eAge) by Horvath's clock on Day 20 of rejuvenated cells (T_{RJ}) compared with non-rejuvenated control cells (T_{CT}) **(B).** Fold change expansion curve of T_{RJ} compared with non-rejuvenated controls **(C).** For reference: Average fold difference between T_{RJ} and T_{CT} at Day 25 was 255.6 vs 14.0, respectively.

Abstract

The T-cell identity and age determine function and fitness over a T-cell's lifespan.[1,2] This is particularly relevant when T cells are derived from patients with chronic viral infection or cancer. It is well known that increased T-cell age and differentiation and increased effector and exhausted phenotypes are associated with reduced antitumor efficacy and the need for higher infusion T-cell numbers for the treatment of hematological or solid tumors during adoptive cell therapies (ACT)[3,4].

In an effort to overcome these barriers, methods to de-differentiate T cells into induced pluripotent stem cells (iPSCs) that return to embryonic immaturity, but lose their functional identity, have been extensively explored in the past years. Early work revealed several challenges to re-differentiate iPSCs into T cells with the desired functional phenotype, requiring a complex and time-consuming process.[5-7] Here, we bring our novel strategy that counters the impact of aging on T-cell function through cellular rejuvenation without de-differentiating to iPSCs. We achieved T-cell rejuvenation via partial reprogramming of aged T cells by transiently expressing transcription factors associated with iPSC reprogramming. This proprietary partial reprogramming methodology reduces epigenetic age and rejuvenates T cells while maintaining the phenotype and function of conventional T cells.

We were the first to illustrate the ability to reduce the epigenetic age of T cells without fully de-differentiating to iPSCs. Our initial studies with PBMCs showed a significant reduction in epigenetic age. On subsequent RNAseq analyses, we observed that rejuvenated and conventional T cells have comparable transcriptomes, suggesting the maintenance of identity. Functionally, however, the T_{RJ} cells are characterized by greatly improved cell-expansion capacity, together with increased expression of markers associated with T-cell stemness, including CCR7 and CD62L. In vitro studies of NY-ESO-1-targeted T-cell receptor (TCR) or a CD19-targeted chimeric antigen receptor (CAR) T_{RJ} cells exhibited improved antitumor properties compared with non-rejuvenated T-cell control (T_{CT}) cells in sequential cell-killing assays. We also confirmed the enhanced in vivo antitumor efficiency of NY-ESO-1 TCR T_{RJ} cells in a murine xenograft tumor model. When tumor-infiltrating lymphocytes (TIL) are rejuvenated with the same rejuvenation technology the TIL showed enhanced cell-expansion capacity, and improvements in T-cell stemness phenotype. These results suggest the potential application of T-cell rejuvenation across multiple adoptive T-cell therapeutic modalities aimed at improving outcomes in patients with solid tumors.

Results (continued)

Expression of reprogramming factors in rejuvenated T cells is transient

Single-cell RNA-seq analysis of T_{CT} and T_{RJ} cells from 4 healthy donors 50 to 55 years of age showed that rejuvenated T cells expressed OSKM factors (Oct4, Sox2, Klf4, and c-Myc) on Day 7, but expression disappeared prior to Day 13 (**Figure 3**). Critically, this non-integrative, transient expression of reprogramming factors mitigates the risk of generating immortalized cells.

Figure 3. RNA-seq analysis of transient expression of OSKM factors in rejuvenated T cells

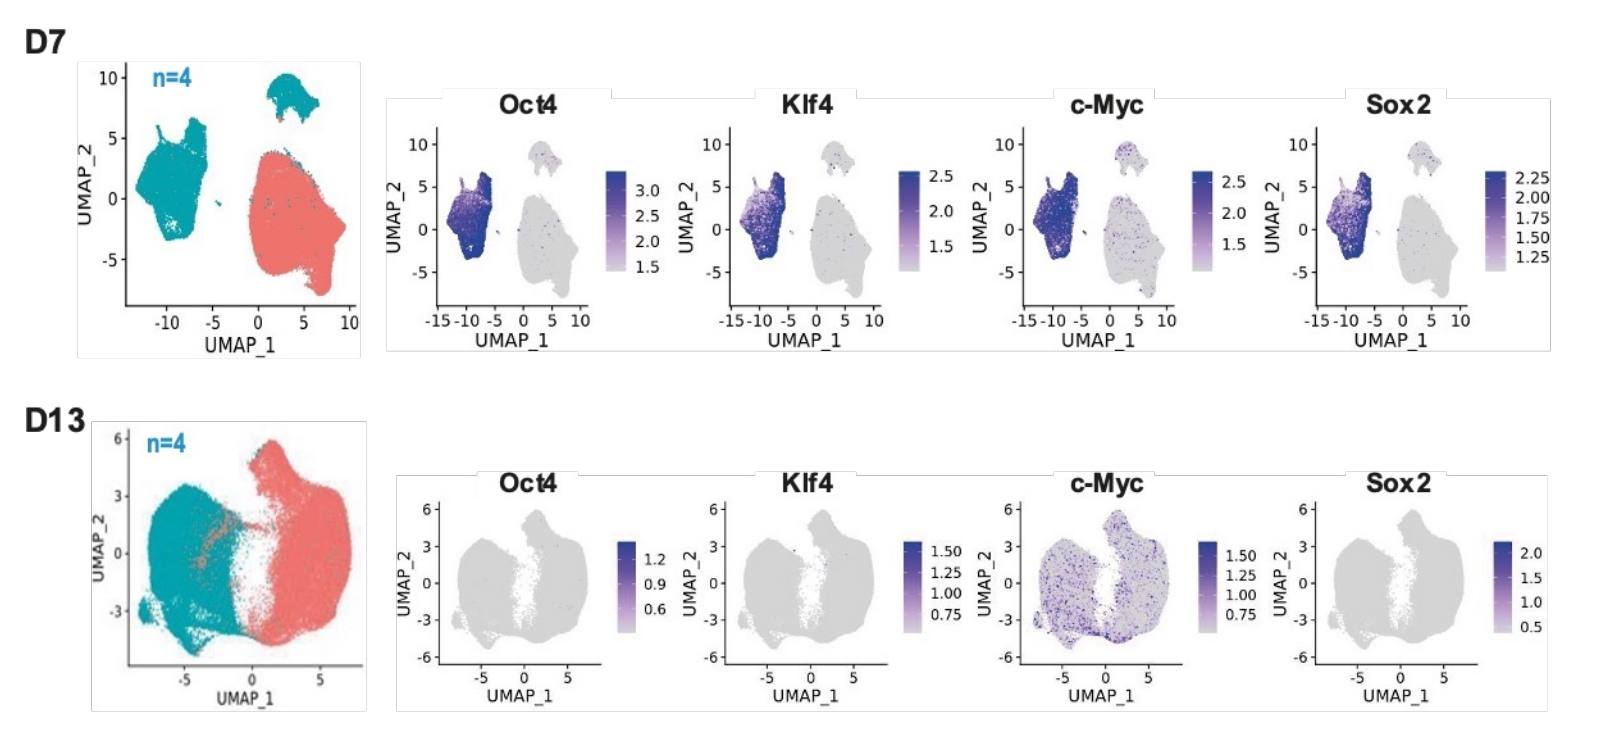
D7
D13
n=4
Oct4
Klf4
c-Myc
Sox2
UMAP_1
UMAP_2
TCT
TRJ

Note: Expression of c-Myc on Day 13 was endogenous.

Rejuvenation produced a conventional T-cell phenotype with improved stemness and metabolism

After the redirection phase, RNA-seq analysis of T_{CT} and T_{RJ} cells from healthy donors 50 to 55 years of age showed that the T_{RJ} cells re-express T-cell–associated genes and compared with controls:

- Did not exhibit abnormal expression of unconventional T-, NK-, or B-cell markers typically expressed by human iPSC-derived T cells (**Figure 4A**).
- Were enriched for expression of genes associated with glycolysis and oxidative phosphorylation at Day 7 and Day 13 compared with control cells, indicating enhanced metabolism (**Figure 4B**).
- Exhibited higher expression of naïve-associated markers characteristic of more stem-like T-cell populations (**Figure 4C**).

Figure 4. Rejuvenated T cells demonstrated a conventional T-cell phenotype with improved metabolism and stemness based on transcriptomic analyses

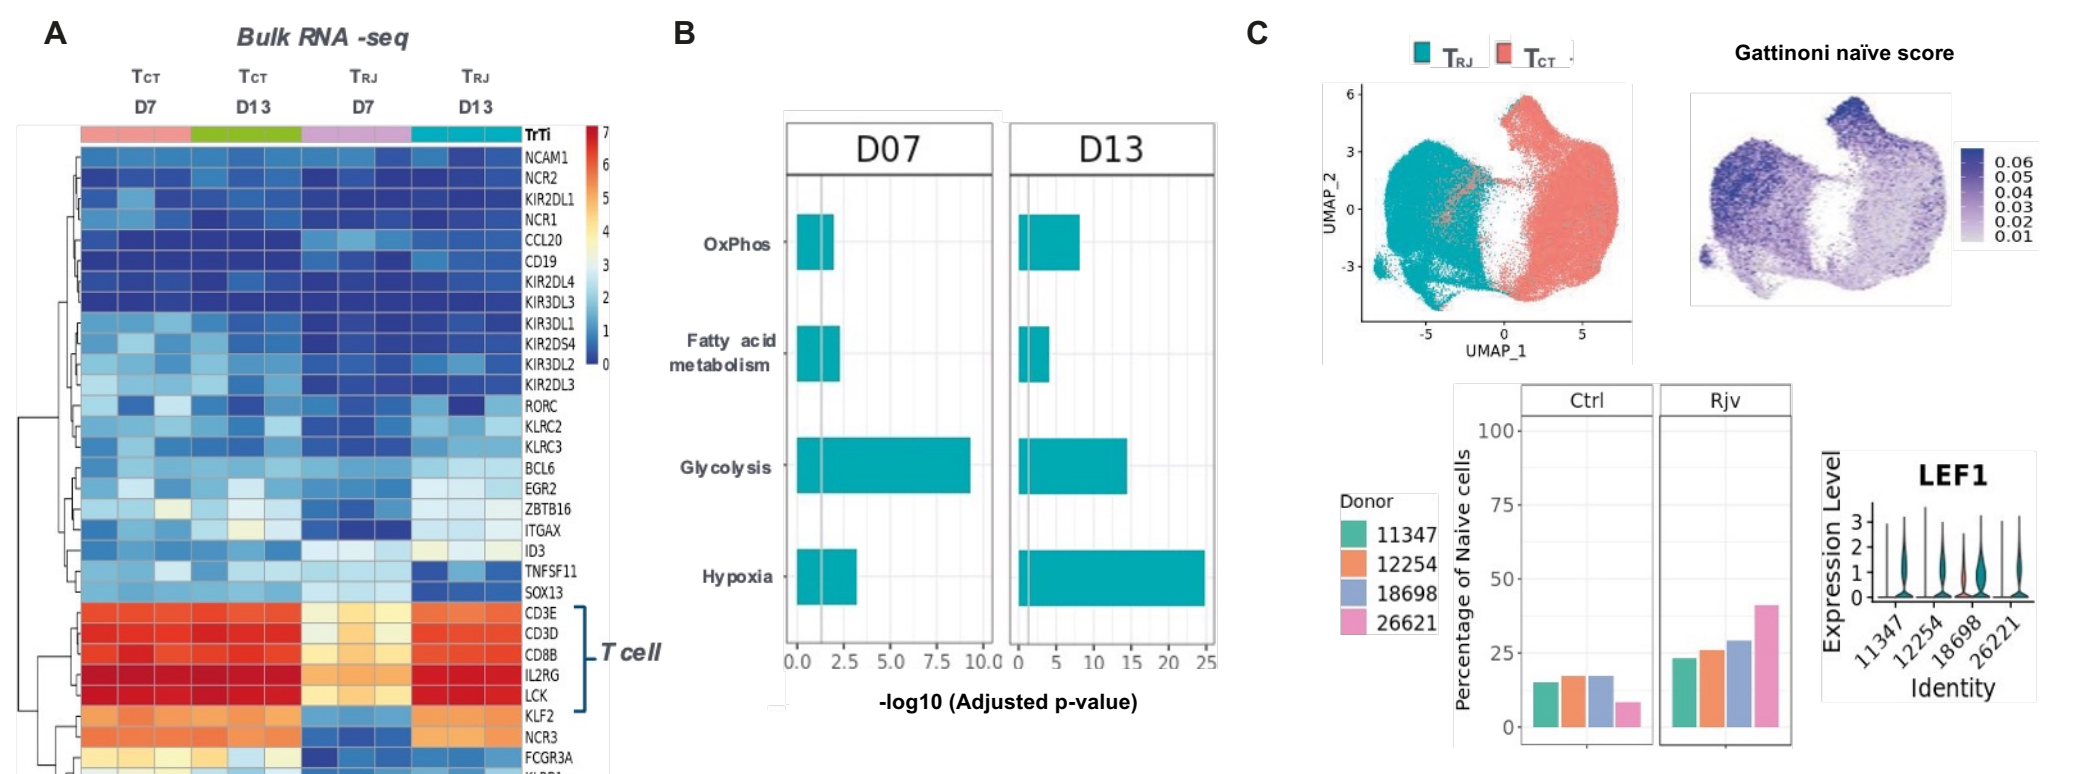
A
Bulk RNA -seq
T cell
B
D07
D13
OxPhos
Fatty acid metabolism
Glycolysis
Hypoxia
-log10 (Adjusted p-value)
C
Gattinoni naïve score
Percentage of Naive cells
Ctrl
Rjv
LEF1
Expression Level
Identity
Donor
11347
12254
18698
26621

(A) Bulk RNA-seq heatmap analysis of previously identified[6] conventional and unconventional genes related to T-cell identity. **(B)** Bulk RNA-seq showed enrichment of DEGs associated with key metabolic features in T_{RJ} at Day 7 and Day 13 compared with T_{CT} cells. **(C)** Single-cell RNA-seq showed elevated expression of naïve-associated markers in rejuvenated cells (T_{RJ}) vs control cells (T_{CT}).

Rejuvenation enhanced the properties associated with T-cell functionality of engineered T-cell therapies and improved their antitumor efficacy

Rejuvenation was evaluated in two models of engineered T-cell therapies: $CD4^+$ and $CD8^+$ NY-ESO-1 TCR T cells and CD19-targeted CAR T cells. Rejuvenation of these models resulted in (**Figure 5**):

- Enhanced proliferation
- Decreased epigenetic age
- Increased stem-like features
- Retained T-cell function and antigen specificity

Sequential killing assays were used to evaluate the cytotoxic function of engineered rejuvenated cells, demonstrating greater persistent antitumor efficacy compared with conventional NY-ESO-1 TCR T cells or CD19 CAR T cells.

Additionally, in an in vivo tumor model, rejuvenated NY-ESO-1 TCR T cells showed improved tumor volume and probability of survival compared with control NY-ESO-1 TCR T cells (**Figure 6**).

Figure 5. Rejuvenation enhanced the functional properties of $CD4^+$ and $CD8^+$ NY-ESO-1 TCR T cells and CD19 CAR T cells and improved their antitumor efficacy

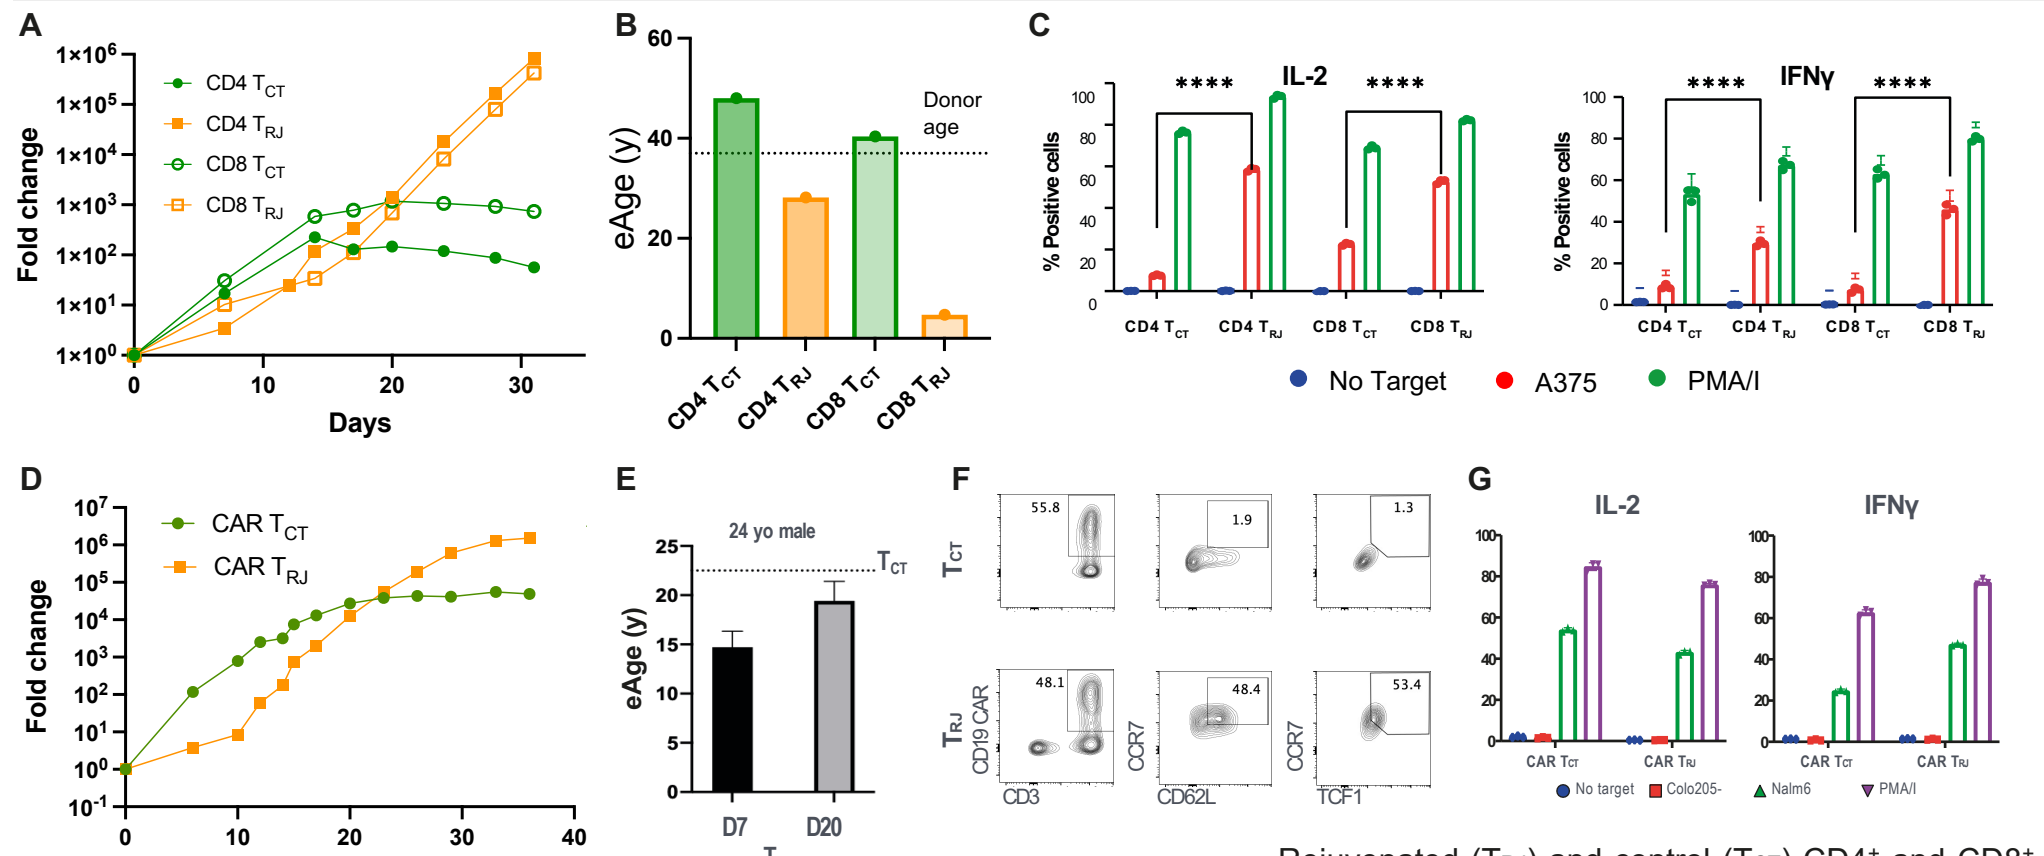
A
Fold change
Days
B
eAge (y)
Donor age
C
IL-2
IFNγ
% Positive cells
No Target
A375
PMA/I
D
CAR TCT
CAR TRJ
E
24 yo male
F
G

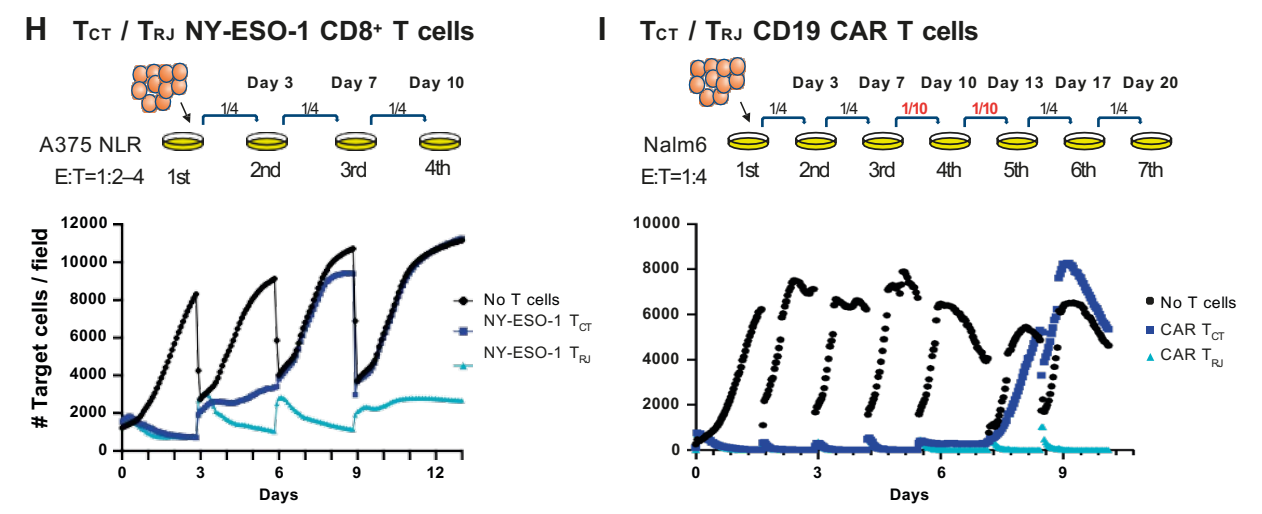
H TCT / TRJ NY-ESO-1 CD8+ T cells
A375 NLR
E:T=1:2–4
I TCT / TRJ CD19 CAR T cells
Nalm6
E:T=1:4
Target cells / field
Days

Rejuvenated (T_{RJ}) and control (T_{CT}) $CD4^+$ and $CD8^+$ NY-ESO-1 TCR T cells of a 37yo healthy donor were evaluated to compare **(A)** cell proliferation, **(B)** epigenetic age, and **(C)** T-cell function and antigen specificity. A375: NY-ESO-1 tumor expressing cell line.

Rejuvenated (T_{RJ}) and control (T_{CT}) CD19 CAR T cells from a 24 yo healthy donor were evaluated to compare **(D)** cell proliferation, **(E)** epigenetic age, **(F)** stemness phenotype, and **(G)** T-cell function and antigen specificity. Nalm6: CD19 tumor expressing cell line. Sequential killing assays of **(H)** rejuvenated NY-ESO-1 TCR T cells (T_{RJ}) and **(I)** rejuvenated CD19 CAR T cells (T_{RJ}) compared with controls (T_{CT}).

Figure 6. In vivo NY-ESO-1 TCR T cells showed enhanced antitumor effects

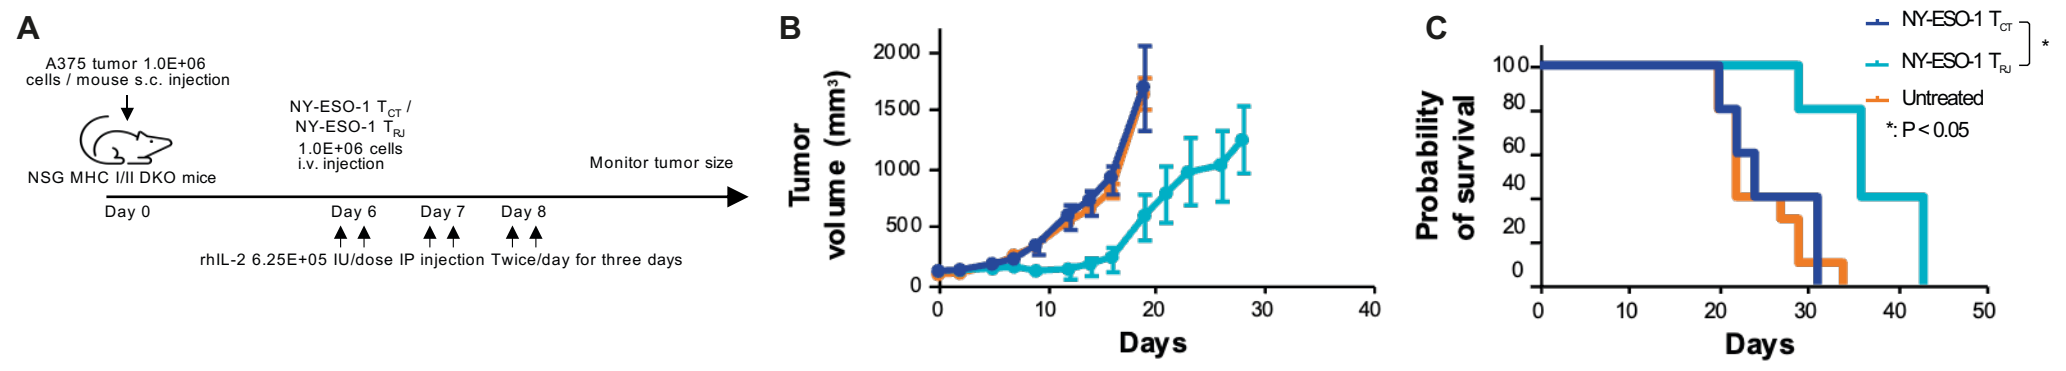
A
A375 tumor 1.0E+06 cells / mouse s.c. injection
NSG MHC I/II DKO mice
Monitor tumor size
rhIL-2 6.25E+05 IU/dose IP injection Twice/day for three days
B
Tumor volume (mm3)
Days
C
Probability of survival
Untreated
*: P < 0.05

(A) Tumor treatment schema. NSG MHC I/II DKO mice were injected with 1.0E+06 A375 tumor cells subcutaneously. ACT was given on Day 6. In each tumor setting, 5–10 mice were included in groups receiving PBS (orange), 1.0E+06 NY-ESO-1 T (dark blue), or 1.0E+06 NY-ESO-1 T (light blue). All mice were given 6.25E+05 IU of rhIL-2 after ACT twice daily for 3 days. Tumor volumes were assessed every two to three days. **(B)** Tumor growth curve. Error bars indicate the mean ± SEM. **(C)** Survival curve. Survival was assessed by a log-rank test.

Results (continued)

Rejuvenation enhanced properties associated with T-cell functionality of TIL

Application of rejuvenation to TIL derived from metastatic melanoma showed improved cell proliferation and stem-like properties as well as a maintained Simpson clonality and TCR repertoire compared with control TIL (**Figure 7**). Similarly, rejuvenated TIL derived from epithelial cancer demonstrated enhanced proliferative capacity and stem-like properties compared with control TIL (**Figure 8**).

Figure 7. Rejuvenation improved cell proliferation capacity, stemness phenotype, and epigenetic age while maintaining the $CD4^+/CD8^+$ population and TCR repertoire in metastatic melanoma TIL

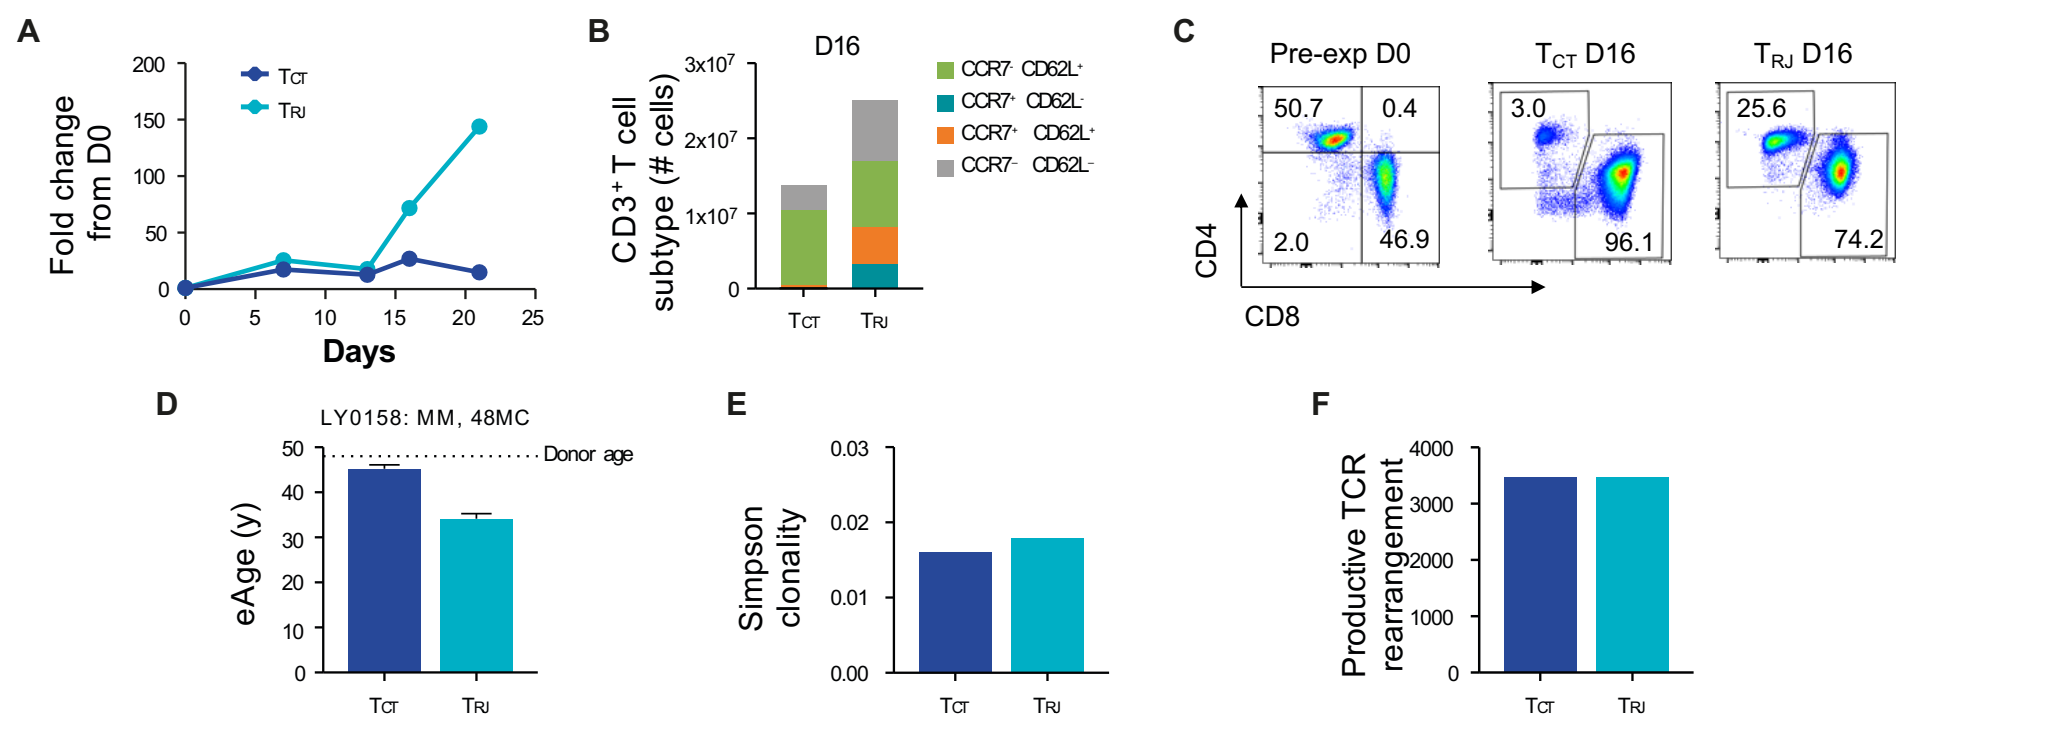
A
Fold change from D0
Days
B
D16
CD3+ T cell subtype (# cells)
C
Pre-exp D0
TCT D16
TRJ D16
CD4
CD8
D
LY0158: MM, 48MC
eAge (y)
Donor age
E
Simpson clonality
F
Productive TCR rearrangement

Rejuvenated (T_{RJ}) and control (T_{CT}) TIL from a 48 yo patient with metastatic melanoma were evaluated to compare **(A)** cell proliferation, **(B)** stemness phenotype, **(C)** preservation of $CD4^+$ and $CD8^+$ populations, **(D)** epigenetic age, **(E)** Simpson clonality and **(F)** number of TCR rearrangements.

Figure 8. Phenotypic and proliferative analysis of rejuvenated TIL from an epithelial cancer

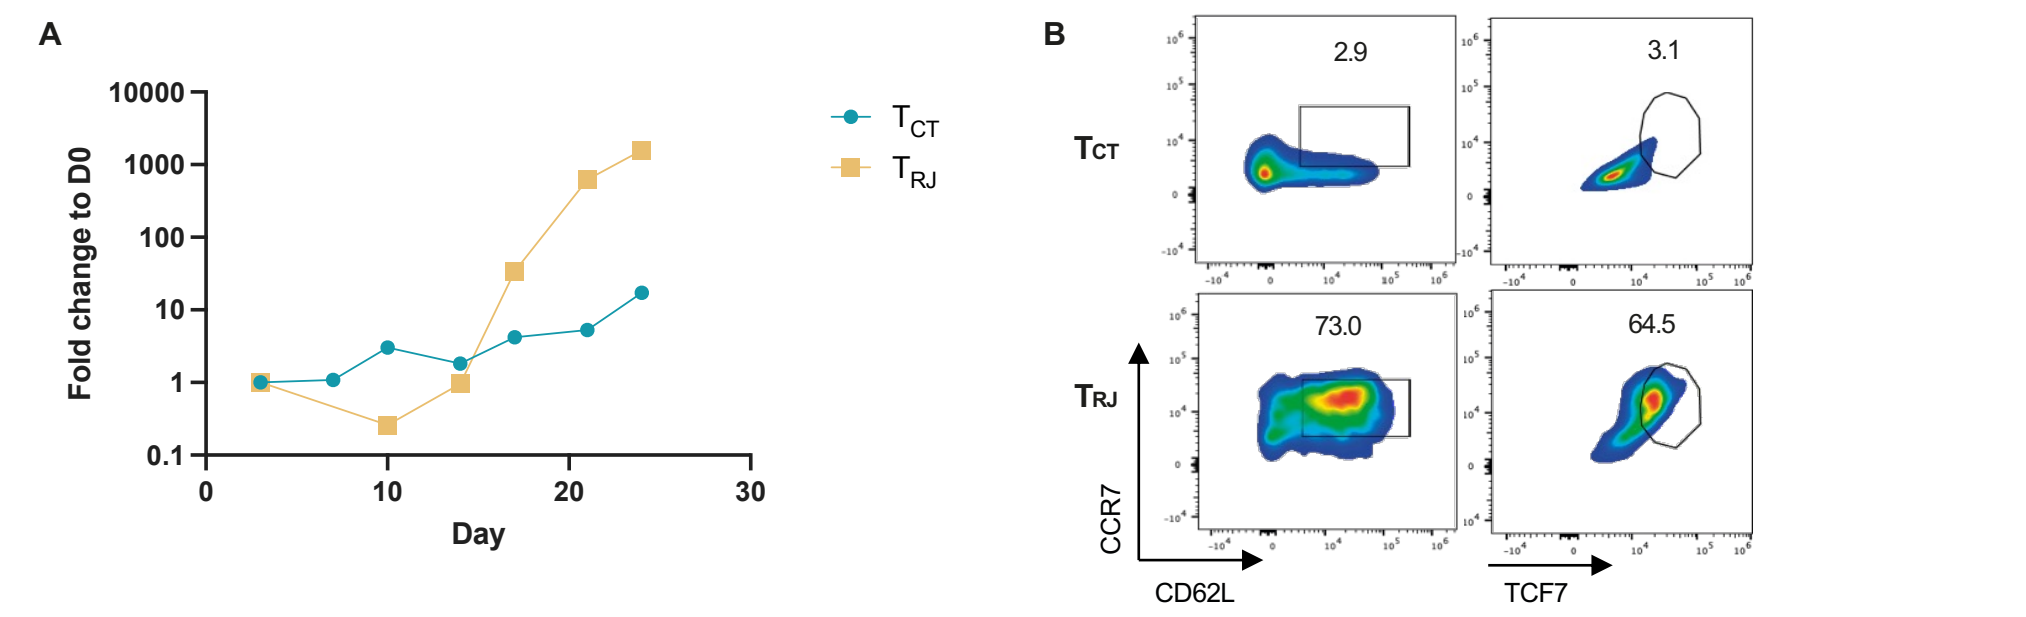
A
Fold change to D0
Day
B
TCT
TRJ
CCR7
CD62L
TCF7

Rejuvenated (T_{RJ}) and control (T_{CT}) TIL from a 66 yo patient with lung cancer were evaluated to compare **(A)** cell proliferation and **(B)** stemness phenotype. For reference: Average fold difference between T_{RJ} and T_{CT} at Day 25 was 1580.0 vs 17.2, respectively.

Conclusion

- Lyell's T-cell rejuvenation technology utilizes partial reprogramming to produce T cells characterized by epigenetic youth, enhanced cell proliferation, improved metabolism, and higher expression of stemness biomarkers. Further research can help elucidate the full scope of benefits of this technology in terms of their capacity for tumor antigen–specific polyclonality, long-term engraftment, and solid tumor eradication in vivo.
- Application of this technology has demonstrated improvements in engineered adoptive T-cell products; in vitro sequential cell killing assays revealed that rejuvenated NY-ESO-1–targeted TCR and CD19-targeted CAR T cells exhibit improved antitumor properties compared with non-rejuvenated T-cell controls. Rejuvenated NY-ESO-1 TCR T cells also show better tumor suppression and survival than control in an in vivo setting. Early application with TIL products yielded similar results, indicating potential utility across several T-cell therapy modalities.
- Lyell's T-cell rejuvenation technology is being advanced for applications in cancer cellular therapy. Through partial reprogramming, Lyell's T-cell rejuvenation technology has the potential to transform conventional T-cell immunotherapies and improve outcomes for patients with solid tumors.

Abbreviations

ACT, adoptive cell therapy; CAR, chimeric antigen receptor; CCR7, C-C motif chemokine receptor 7; CD, cluster of differentiation; c-Myc, MYC proto-oncogene; DEG, differentially expressed gene; eAge, epigenetic age; E:T, effector to target ratio; IFNγ, interferon gamma; IL-2, interleukin-2; IP, intraperitoneal; iPSC, induced pluripotent stem cells; IU, international units; Klf4, Krüppel-like factor 4; MM, multiple myeloma; NK, natural killer; NY-ESO-1, New York esophageal squamous cell carcinoma 1; Oct4, octamer-binding transcription factor 4; OSKM factors, Oct4, Sox2, Klf4, and c-Myc; OxPhos, oxidative phosphorylation; PBS, phosphate-buffered saline; RNA-seq, RNA sequencing; SC, subcutaneous; SEM, standard error of the mean; Sox2, SRY-box transcription factor 2; TCF1, T-cell factor 1 (encoded by TCF7); TCF7, transcription factor 7; TCR, T-cell receptor; T_{CT}, control T cell; TIL, tumor-infiltrating lymphocytes; T_{RJ}, rejuvenated T cell; y, years; yo, year old.

References

1. Crompton JG, et al. *Trends Immunol.* 2014;35(4):178–185.
2. Crompton JG, et al. *Cell Stem Cell.* 2013;12(1):6–8.
3. Kishton RJ, et al. *Curr Opin Immunol.* 2022;74:39–45.
4. Nikolich-Zugich et al. *Nat Rev Immunol.* 2008;8(7):512–522.
5. Vizcardo R, et al. *Cell Stem Cell.* 2013;12(1):31–36.
6. Maeda T, et al. *Cancer Res.* 2016;76(23):6839–6850.
7. Vizcardo R, et al. *Cell Rep.* 2018;22(12):3175–3190.

Acknowledgments

This project was fully supported by Lyell Immunopharma, Inc. We would like to thank Lyell's T-cell rejuvenation and BATT teams for their technical support and Luca Gattinoni for critical feedback. We would also like to thank the Clock Foundation for epigenetic clock analyses, Maria Romanova (Molecular House, Inc.) for graphical design support, Melanie Styers (Veracity Science), and Madison Fagan (BOLDSCIENCE, Inc.) for medical writing support.

Presented at AACR Special Conference In Cancer Research: Tumor Immunology and Immunotherapy 2023; Oct 1-4, 2023; Toronto, Canada

©2023 Lyell Immunopharma, Inc