Epi-R™ Technology Produces a Polyclonal TIL Product (LYL845) With a Greater Expansion Success Rate Across Hot and Cold Tumors, Improved Product Phenotype, and Maintenance of TCR Diversity

Yogin Patel, Benjamin D. Harris, Melissa Bedard, Joanna Kritikou, Meri Galindo Casas, Carson Harms, Melissa DeFrancesco, Purnima Sundar, Nicholas P. Restifo, Gary Lee, Shobha Potluri, Suman Kumar Vodnala

Lyell Immunopharma, Inc., South San Francisco, CA and Seattle, WA, USA

Abstract 370

Background

- Adoptive cell therapy (ACT) using tumor-infiltrating lymphocyte (TIL) therapy is a promising method for cancer treatment
- TILs that are highly enriched with tumor-reactive T-cell clones have been shown to mediate treatment response in advanced cancers[1-3]
- Conventional TIL products comprise a mixture of extensively differentiated T cells and low levels of stem-like cells
- Higher proportions of stem-like T cells have been associated with improved outcomes in patients treated with TIL therapies[4]
- Current standard expansion protocols reduce TIL stemness and TCR diversity through progressive differentiation during *ex vivo* expansion, which can impact treatment efficacy and outcomes[4-5]
- LYL845 is an autologous TIL product enhanced with Epi-R™ technology, a proprietary epigenetic reprogramming protocol designed to improve T-cell stemness and preserve polyclonality

Epi-R: An *Ex Vivo* Epigenetic Reprogramming Technology

- LYL845 was prepared with Epi-R technology designed to intentionally and reproducibly generate TILs with durable stemness that can proliferate, persist, and provide prolonged anti-tumor functionality (**Figure 1**)
- The Epi-R protocol is composed of a specifically formulated cell culture media with optimized cytokine composition and cell activation that induces metabolic reprogramming via reduced glycolysis and hypoxia
- LYL845 was produced at research scale using a total of 17 samples from immunologically hot (melanoma and lung) and cold (colorectal) tumors and was compared with control TIL products generated without Epi-R
- LYL845 was generated from five tumor samples, produced at large scale, and compared with corresponding research-scale products

Figure 1: T-Cell Composition of Epi-R vs. Standard Expansion Protocol T-Cell Preparations

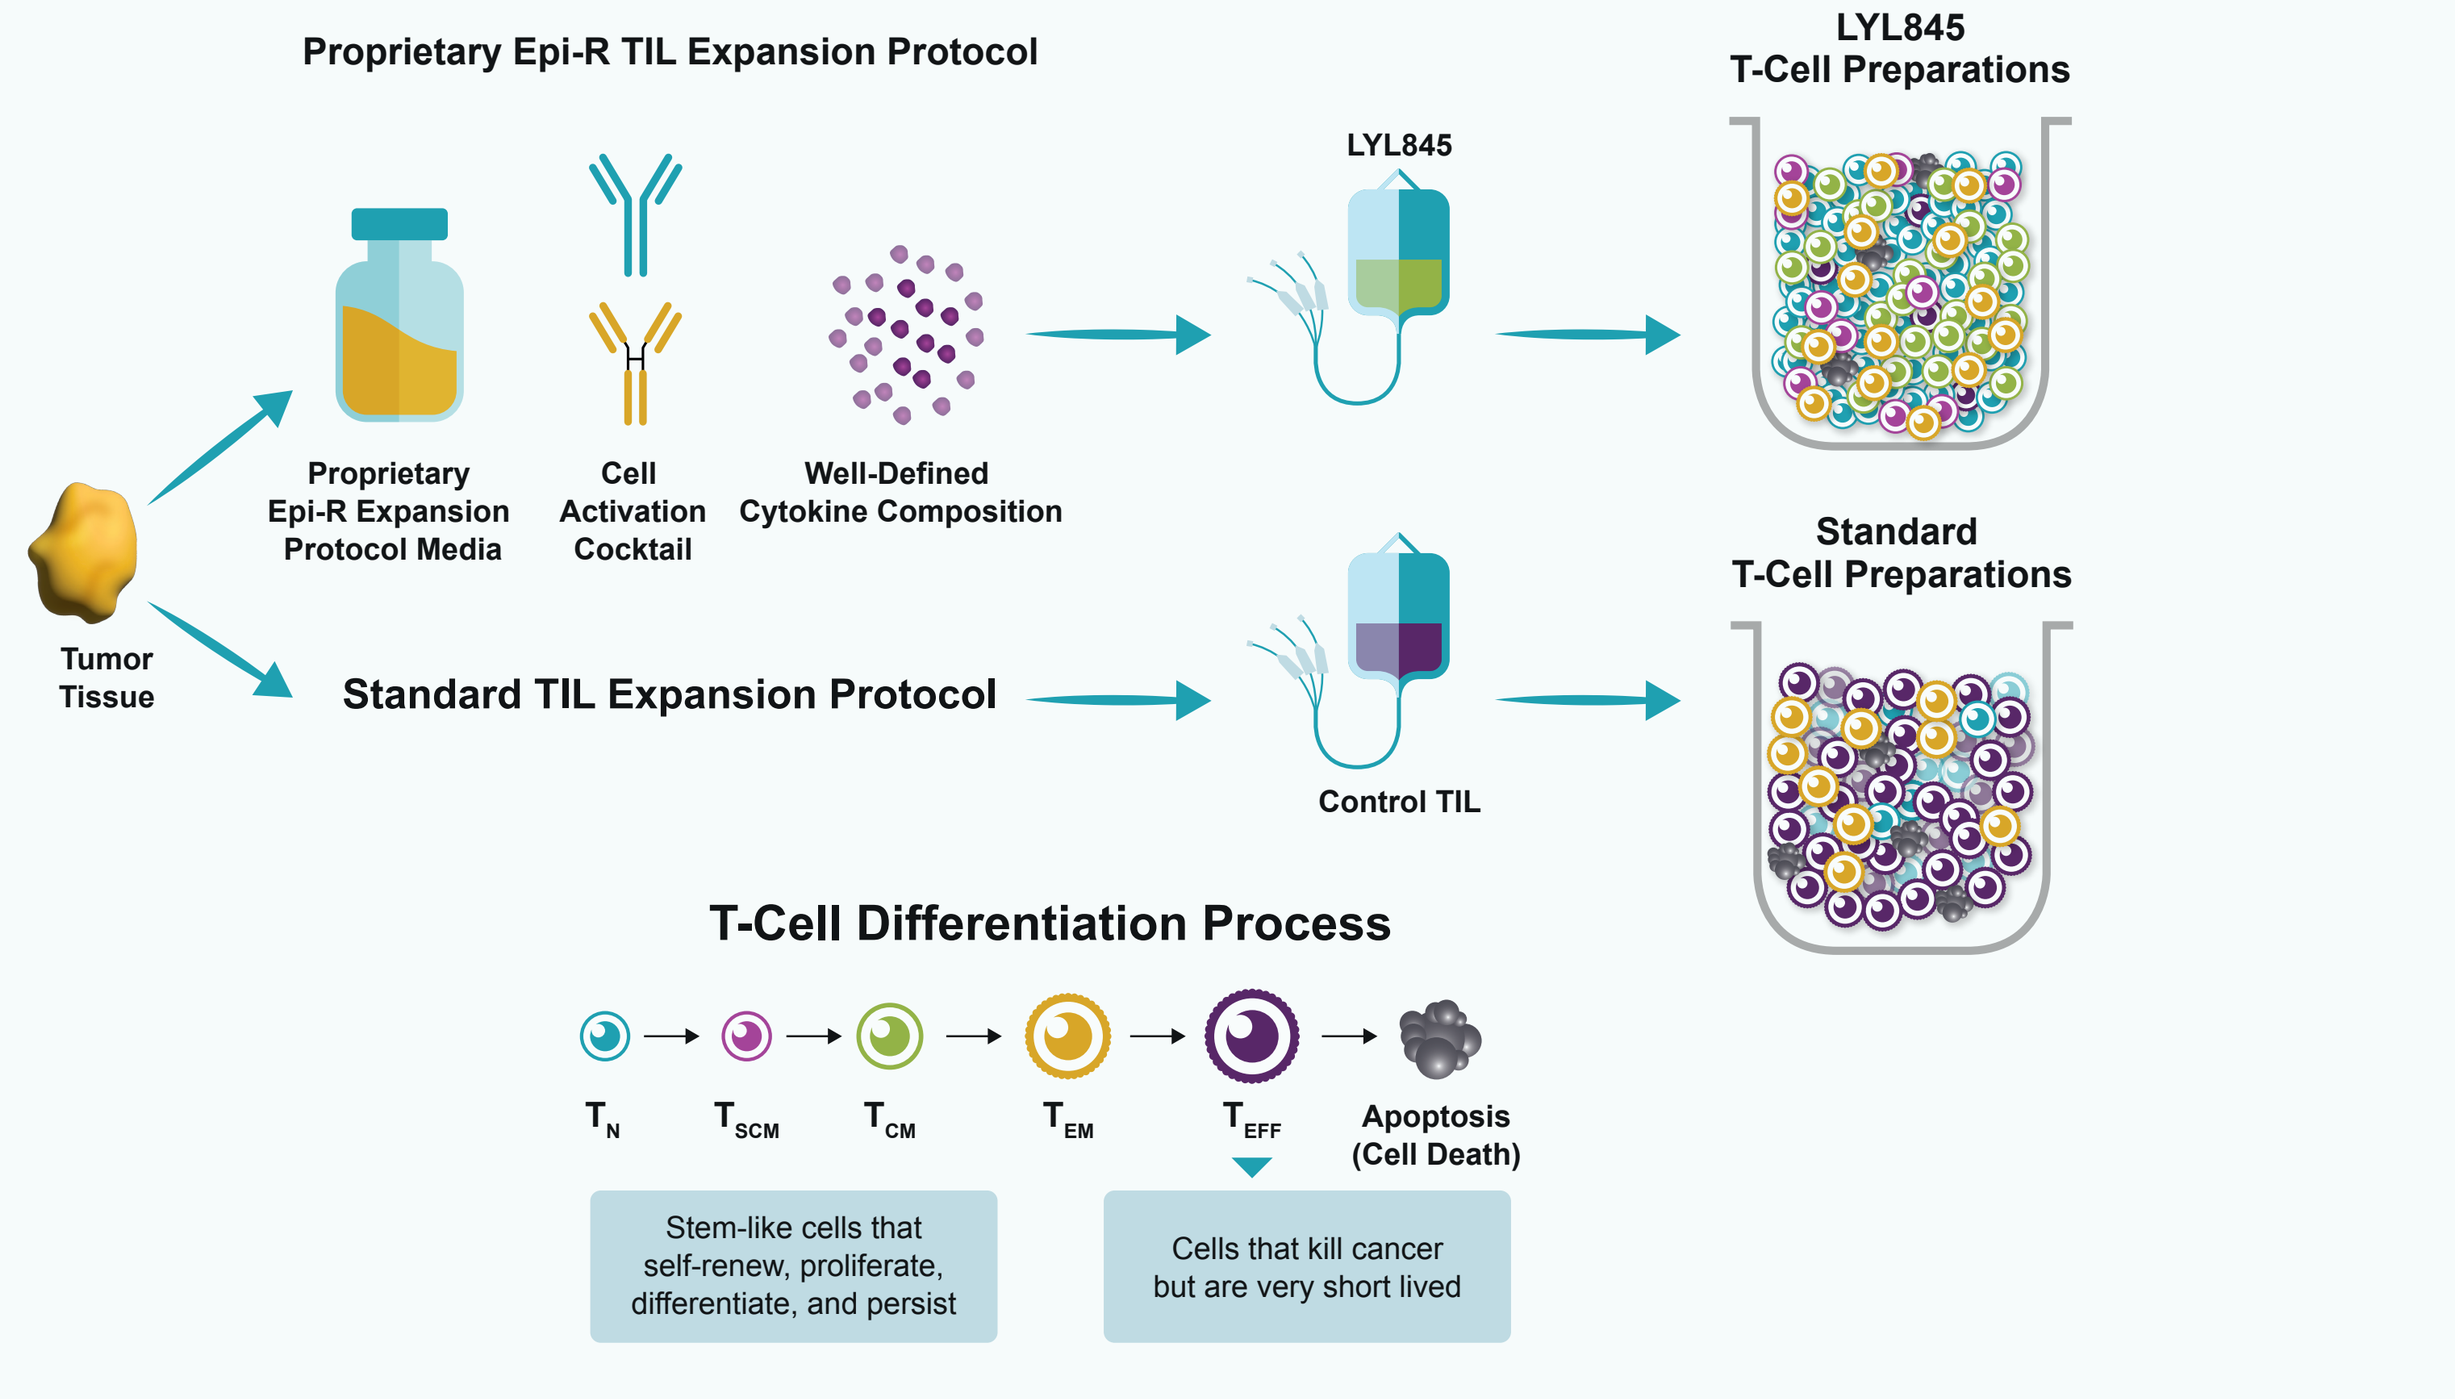
Proprietary Epi-R TIL Expansion Protocol
LYL845 T-Cell Preparations
LYL845
Proprietary Epi-R Expansion Protocol Media
Cell Activation Cocktail
Well-Defined Cytokine Composition
Tumor Tissue
Standard TIL Expansion Protocol
Control TIL
Standard T-Cell Preparations
T-Cell Differentiation Process
T_N → T_{SCM} → T_{CM} → T_{EM} → T_{EFF} → Apoptosis (Cell Death)
Stem-like cells that self-renew, proliferate, differentiate, and persist
Cells that kill cancer but are very short lived

Demonstration of research scale Epi-R TIL expansion protocol. The Epi-R TIL product (LYL845) has a higher proportion of stem-like T cells compared with the control product using a standard expansion protocol.

Methods

LYL845 and Control TIL Manufacturing Methodology

- TIL products were generated from melanoma tissue (with or without prior ICB) and tissues from lung and colorectal cancers
- The Epi-R protocol was used to produce LYL845, and a standard expansion protocol was used to produce control TILs[3,6]

LYL845 and Control TIL Product Assay and Sequencing

- Characteristics of the resulting products (LYL845 and control) were compared using a matrix of assays including:
 - Flow cytometry to measure markers of stemness
 - Bulk RNA-seq to identify differentially expressed genes associated with stemness, exhaustion, and cellular metabolic fitness
 - Bulk TCR Vβ sequencing to assess polyclonality

Results

Key Findings

When compared with a control TIL product, LYL845 demonstrated successful TIL expansion with:

- Enrichment of CD8+ T cells
- Increased proportion of stem-like T cells
- Preservation of high clonal diversity
- Higher proportion of stem-like CD8+ T cells and better expansion in products from samples with prior ICB

These characteristics are maintained in large-scale production

LYL845 Demonstrated Successful TIL Expansion Favoring Enrichment of CD8+ T cells

- Epi-R technology resulted in a 100% success rate of LYL845 expansion across all three tumor types vs a 70% success rate with the control process (**Figure 2A**)
- Epi-R technology enriched the LYL845 product for CD8+ T cells compared with control TILs (**Figure 2B**)

Figure 2: Expansion Success Rate and CD8+ T-Cell Enrichment in the Final TIL Products

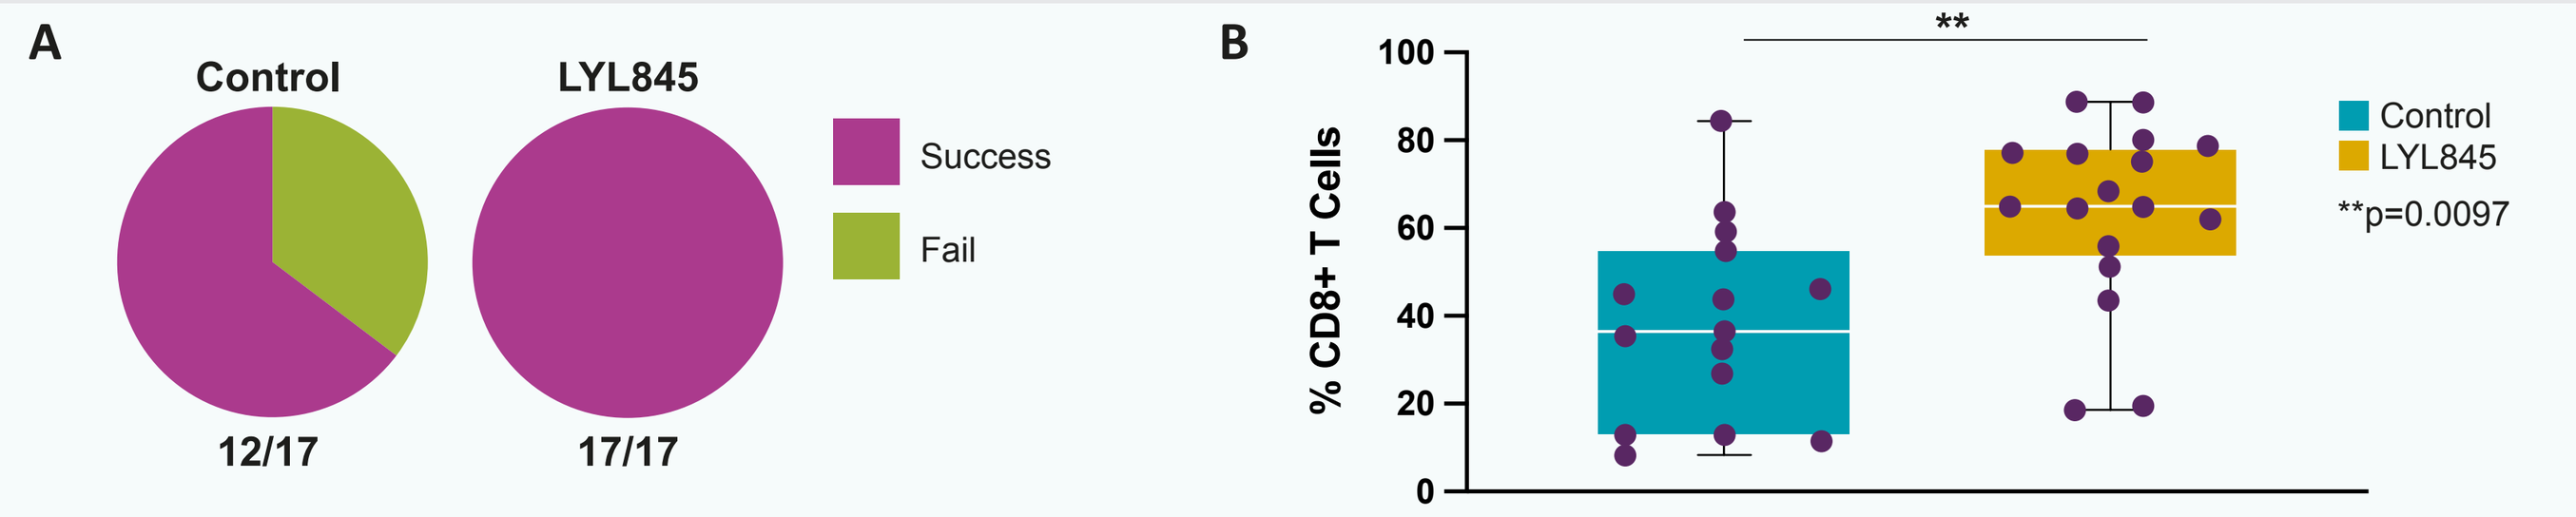
A
Control
LYL845
Success
Fail
12/17
17/17
B
% CD8+ T Cells
Control
LYL845
**p=0.0097

(**A**) A 1.5-fold expansion was achieved at the end of pre-REP and continued to REP for all 17 tumors undergoing Epi-R production, while only 12 tumors met the minimum fold expansion criterion to continue to REP for the control production. (**B**) Box plot demonstrating the percentage of CD8+ T cells across all tumor indications.

LYL845 Demonstrated Higher Frequency of Co-Stimulatory Markers Associated With Stemness and Gene Expression Consistent With Stem-Like T Cells

- TIL products enriched with CD8+ T cells that express co-stimulatory markers (CD27, CD62L, and CD127) are associated with T-cell stemness[4,5]
- T cells that are CD39−CD69− are identified as stem-like and have been correlated with positive clinical outcomes, while CD39+CD69+ T cells are highly differentiated and can correlate with poor clinical outcomes[4]
- A gating strategy was used to analyze CD8+ T-cell skewing and stemness phenotypes of the TIL products (**Figure 3**)
- Analysis of LYL845 showed an increase in stem-like T cells with these attributes compared with a control TIL product (**Figures 4, 5**)
- Stemness and metabolic fitness genes were upregulated, while exhaustion genes were down-regulated in LYL845 compared with a control TIL product (**Figure 6**)

Figure 3: Gating Strategy for Identifying Co-Stimulatory Positive and Stem-Like CD8+ and CD4+ T Cells

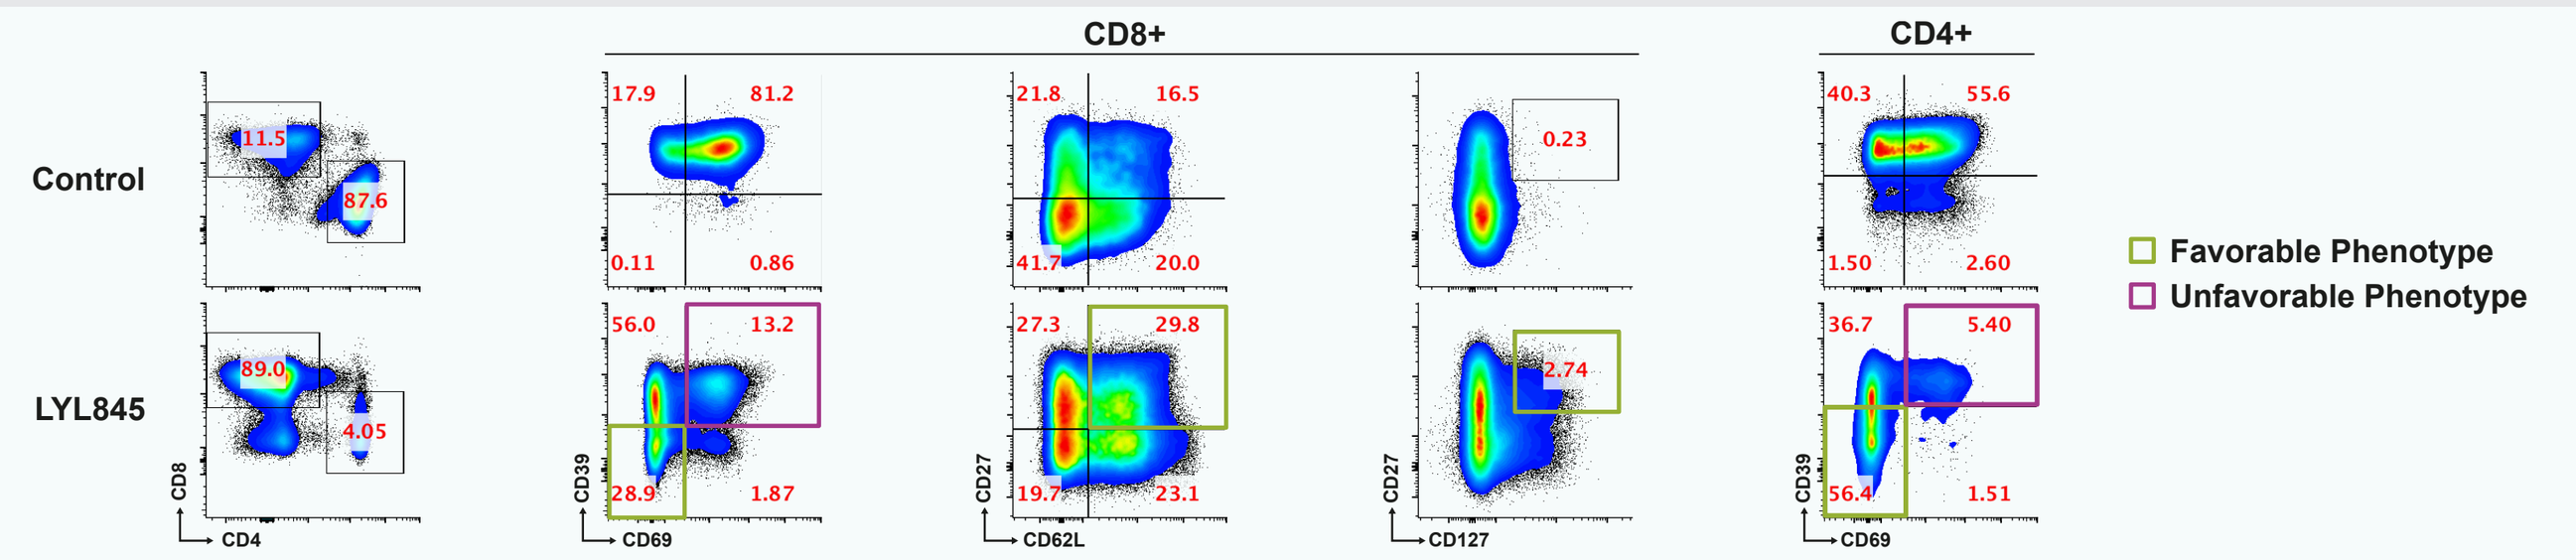
Control
LYL845
CD8+
CD4+
Favorable Phenotype
Unfavorable Phenotype

TIL products were gated on lymphocytes by FSCA vs SSCA, then singlets by FSCA vs FSCH, then live cells by exclusion dye against FSCA, then CD3+ T cells by CD3+ vs FSCA. The CD8+ T cells were further analyzed for stemness characteristics by plotting CD39 against CD69 and gating on the double-positive population (purple, CD39+CD69+) and double-negative population (green, CD39−CD69−). For stem-like, double-positive populations in the CD8+ T-cell subset, CD62L and CD27 or CD27 and CD127 were plotted against each other and gated on the double-positive population (green, CD62L+CD27+ or CD27+CD127+).

Figure 4: The Epi-R Process Enriched Co-Stimulatory Positive CD8+ TILs Compared With the Control Process

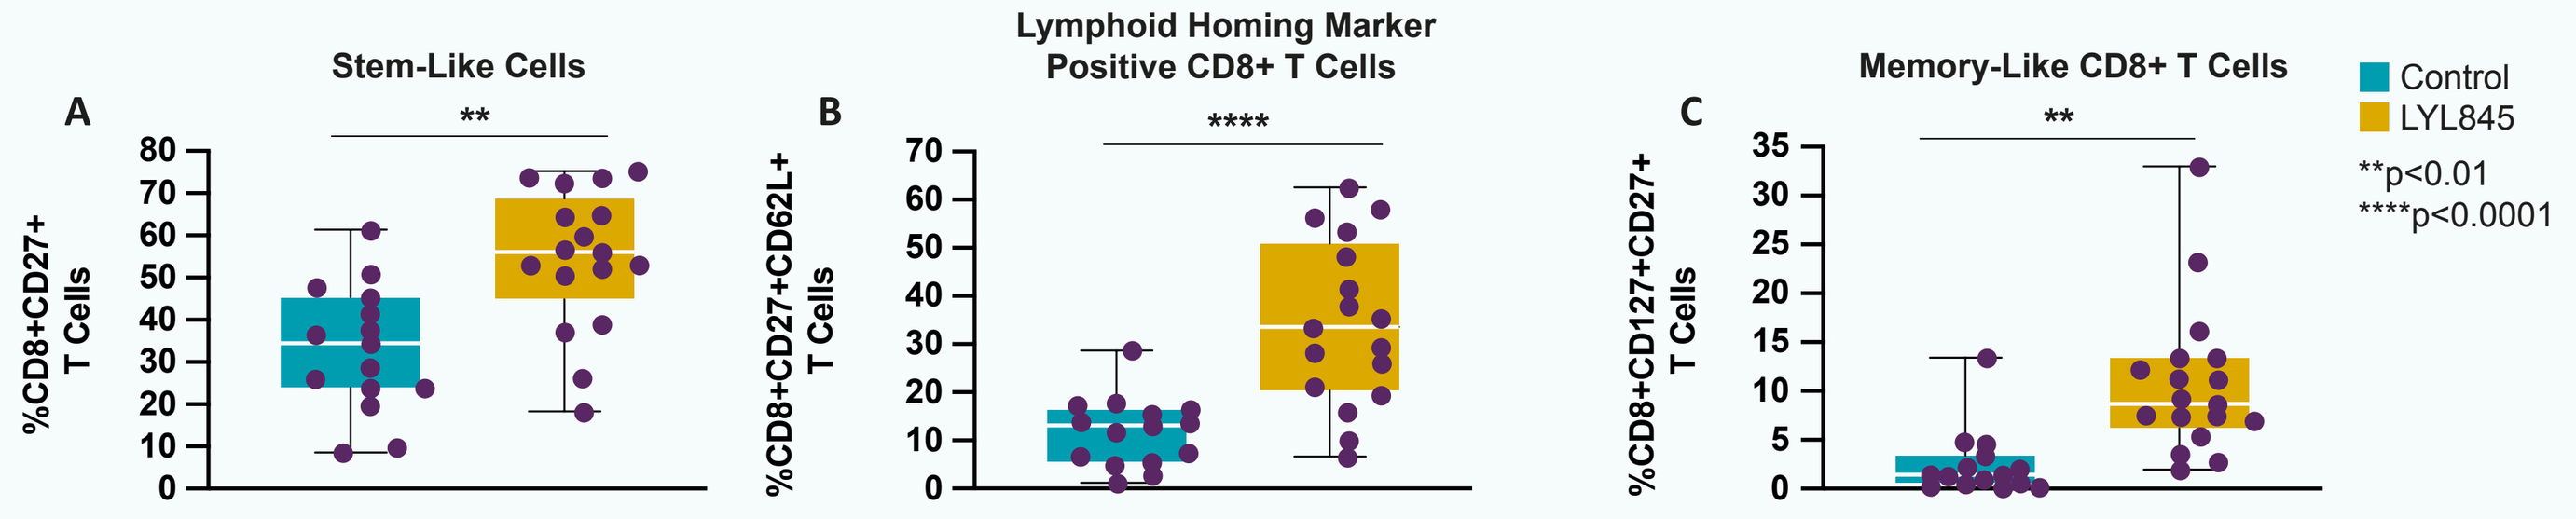
A
Stem-Like Cells
%CD8+CD27+ T Cells
B
Lymphoid Homing Marker Positive CD8+ T Cells
%CD8+CD27+CD62L+ T Cells
C
Memory-Like CD8+ T Cells
%CD8+CD127+CD27+ T Cells
Control
LYL845
**p<0.01
****p<0.0001

Box plots demonstrating (**A**) the frequency of CD8+CD27+ T cells, (**B**) the increased frequencies of CD8+CD27+CD62L+ T cells, and (**C**) CD8+CD127+CD27+ T cells in all tumor indications as compared with control products.

Figure 5: Enrichment of CD39−CD69− Stem-Like CD8+ T Cells in LYL845 vs Control TIL Product

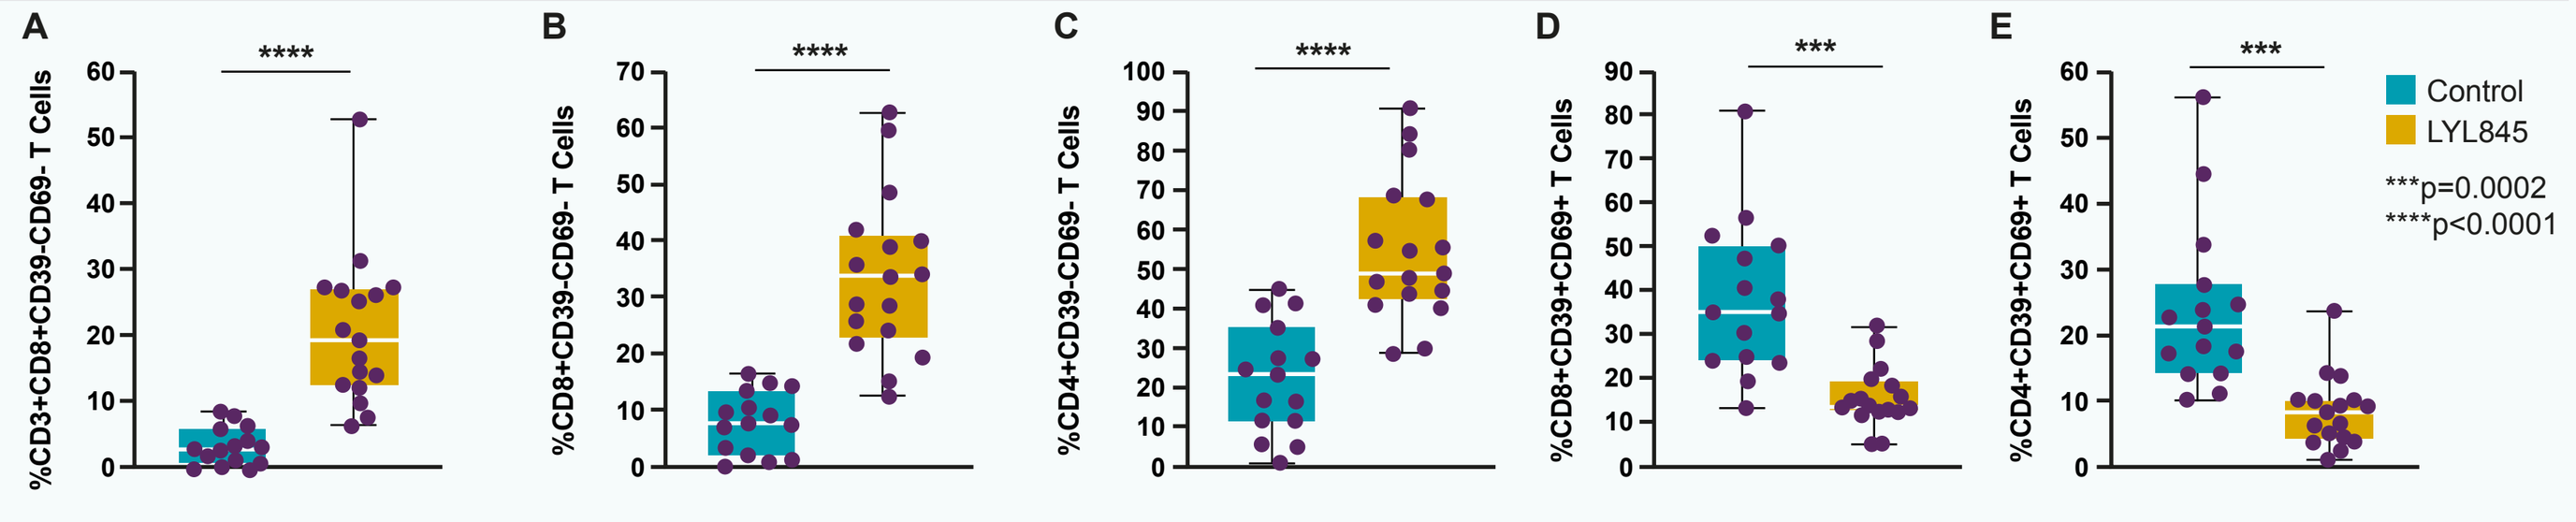
Control
LYL845
***p=0.0002
****p<0.0001

(**A**) Box plot demonstrating percentage of CD3+ cells that were CD8+CD39−CD69− in all tumor indications. Box plots demonstrating (**B**) CD8+ cells that were CD39−CD69− and (**C**) CD4+ T cells that were CD39−CD69− in all tumor indications. (**D**) CD8+ and (**E**) CD4+ T cells were plotted on CD39 vs CD69 and the percentage of the CD39+CD69+ population was calculated in all tumor indications.

Figure 6: LYL845 Was Enriched for Genes Associated With Stem-Like Cells and Enhanced Metabolic Fitness, and Showed a Reduction in Exhaustion-Associated Genes When Compared With the Control TIL Product

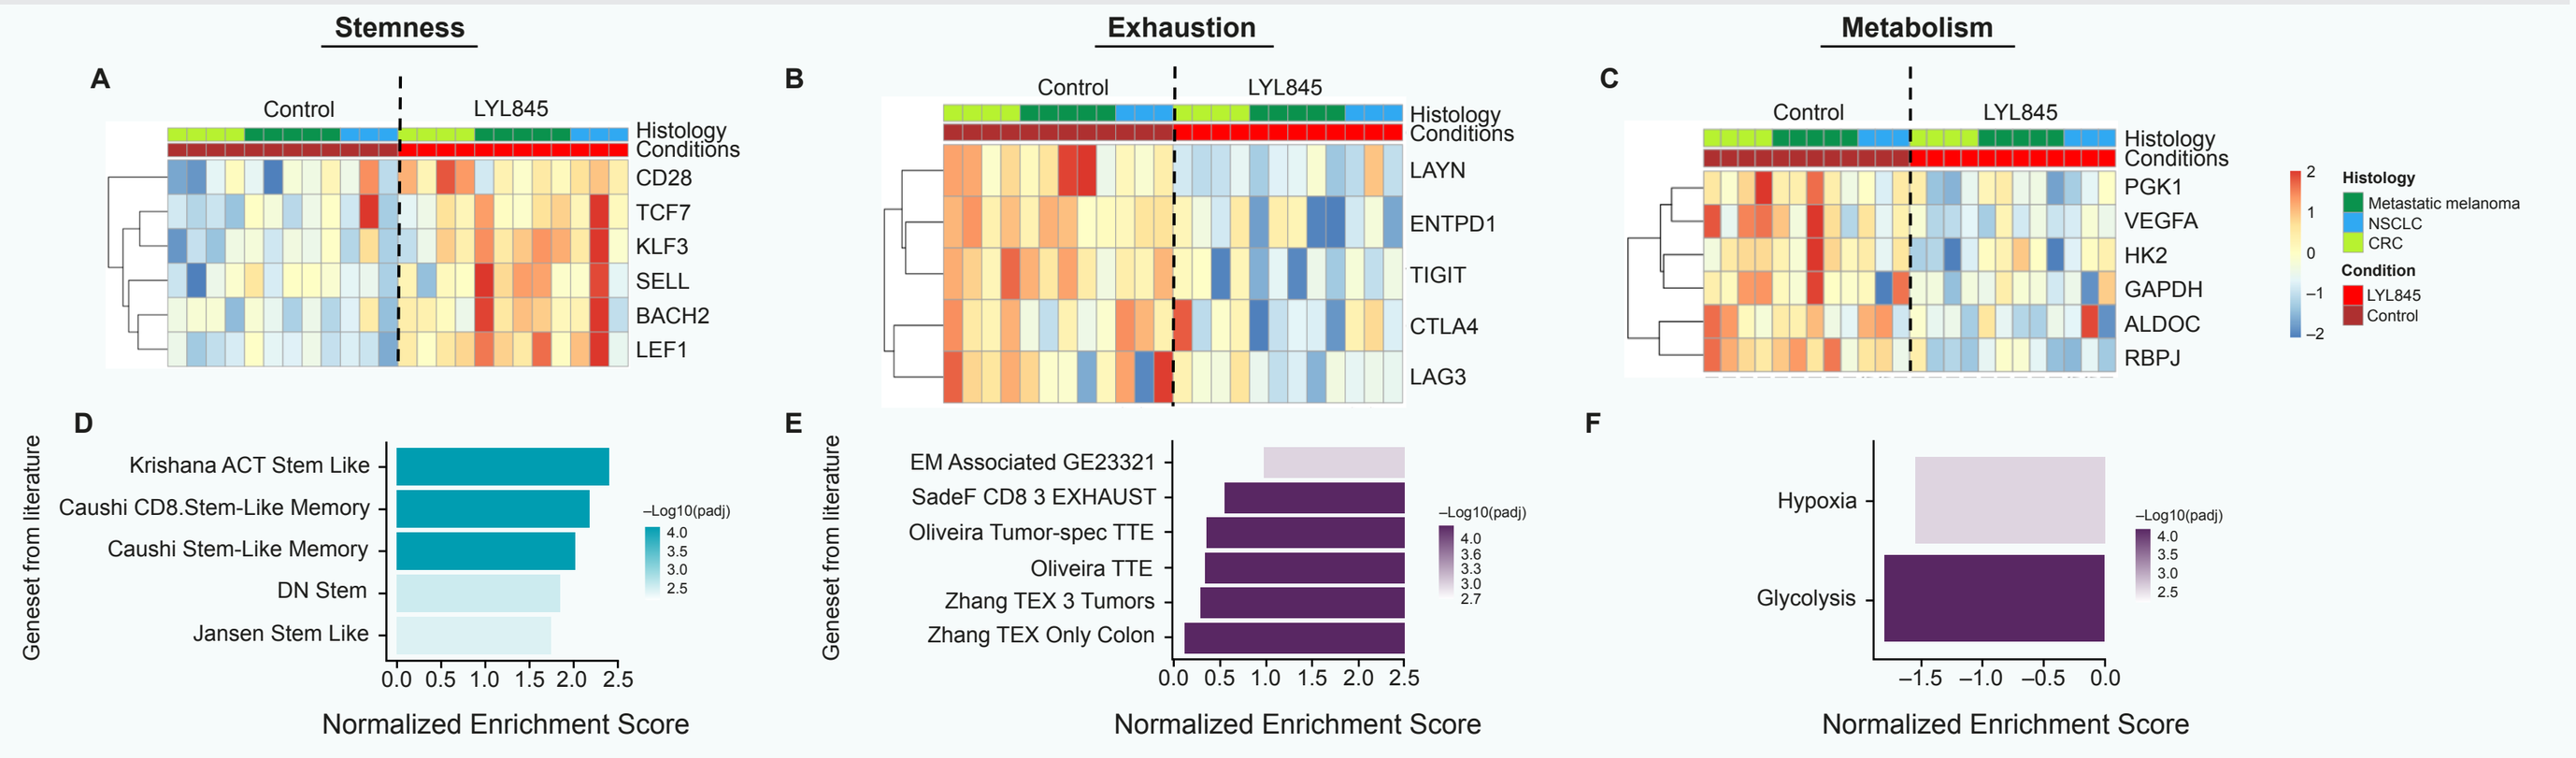
Stemness
Exhaustion
Metabolism
Normalized Enrichment Score

Heatmaps show Z-score expression of differentially expressed genes associated with (**A**) stemness, (**B**) exhaustion, and (**C**) metabolism between Epi-R (LYL845) and control products from 12 donors. Gene set enrichment analysis identifies (**D**) up-regulation of stemlike markers, (**E**) down-regulation of exhaustion markers, and (**F**) down-regulation of hypoxia and glycolysis markers in LYL845 products.

LYL845 Preserved High Clonal Diversity Relative to Source Tumor

- Maintaining the polyclonality of source TILs during *ex vivo* TIL expansion and production is correlated with effective tumor recognition and killing[7]
- Clones tend to be lost during standard *ex vivo* TIL expansion, thus reducing overall clonal diversity in the final TIL product[7]
- TIL products with high clonal diversity have been associated with improved tumor neoantigen recognition and effectiveness[8]
- Polyclonality was preserved in LYL845 as measured by the Simpson Clonality index using bulk TCR Vβ sequencing data (**Figure 7**)

Figure 7: LYL845 Polyclonality Preserved as Measured by Simpson Clonality Index

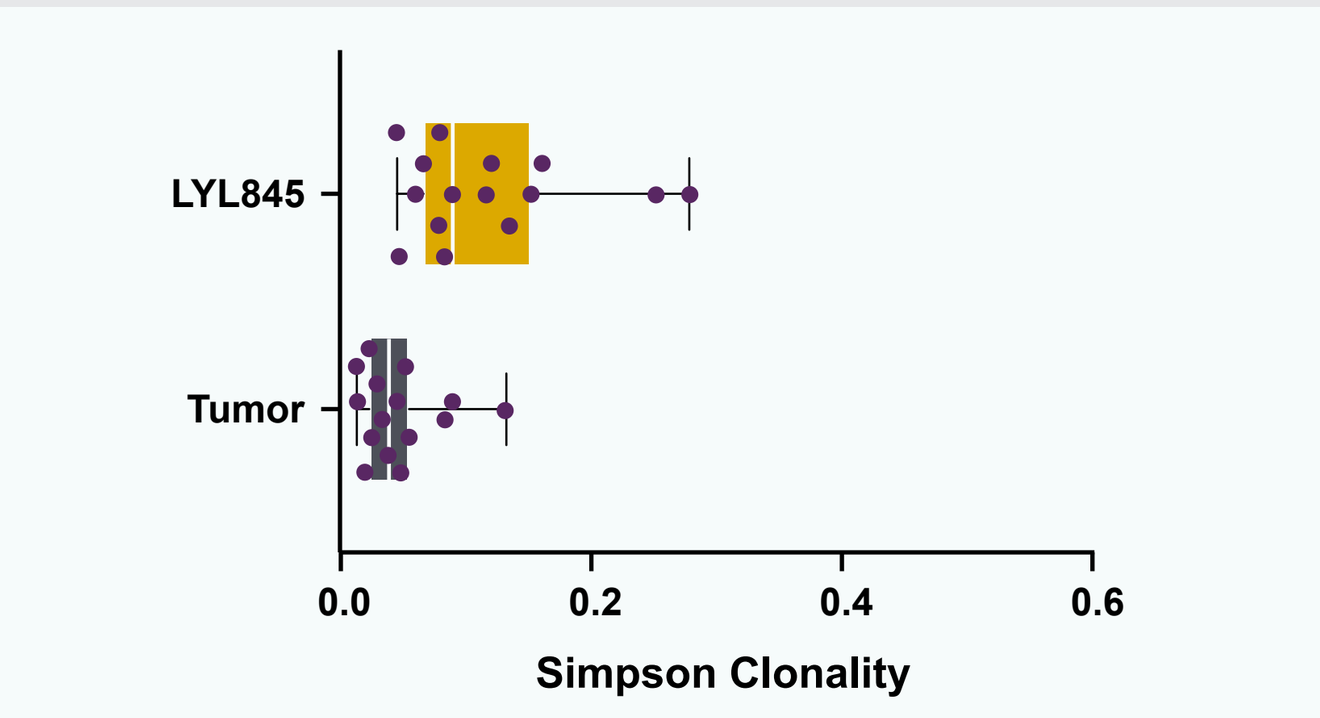
LYL845
Tumor
Simpson Clonality

Boxplot demonstrating Simpson Clonality between initial tumor and LYL845 TIL product.

LYL845 Demonstrated Enrichment of Stem-Like CD8+ T Cells with Prior ICB

- Recent data suggest that disease response in patients with ICB-refractory melanoma is driven by the presence of CD8+ T cells in TIL products[2]
- LYL845 produced from melanoma tissue in patients with ICB-refractory disease had successful expansion using the Epi-R protocol and maintained enrichment of CD8+ T cells with a stem-like phenotype (**Figure 8**)

Figure 8: LYL845 from ICB-refractory Metastatic Melanoma Enriched for CD8+ T Cells with a Stem-Like Phenotype Compared With a ICB-refractory Control TIL Product

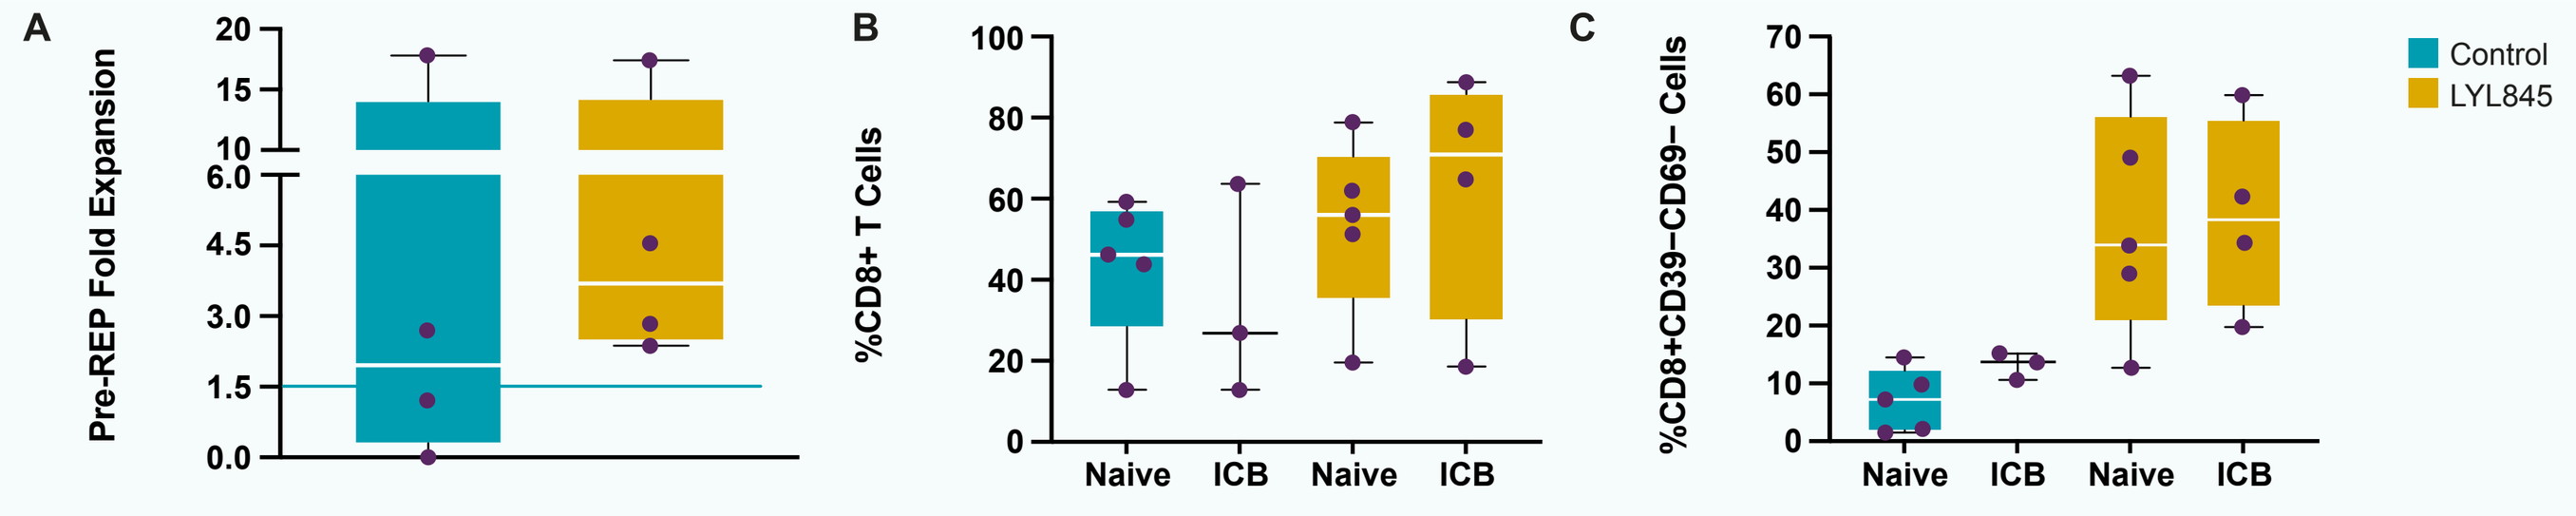
A
Pre-REP Fold Expansion
B
%CD8+ T Cells
Naive
ICB
C
%CD8+CD39−CD69− Cells
Control
LYL845

(**A**) Comparison of TIL expansion in control TIL product and LYL845 produced from ICB-refractory metastatic melanoma. Phenotypic comparison of control and LYL845 TIL product created from naïve and ICB-refractory metastatic melanoma in (**B**) CD8+ T cells and (**C**) CD8+CD39−CD69− cells.

The Epi-R Protocol Maintained Key TIL Attributes in Large-Scale LYL845 Production

- TIL products from four metastatic melanoma tumors and one lung tumor from fresh or frozen single-cell suspensions were expanded using research- and large-scale Epi-R manufacturing, and the resulting TIL products were compared
- The key attributes in research-scale products were reproduced in LYL845 large-scale products (**Figure 9**)
- Large-scale production similarly enriched LYL845 product with CD8+ T cells co-expressing markers of stemness and stimulatory co-receptors
- Large-scale LYL845 production also demonstrated a high retention of polyclonality
- Up-regulation of stemness-associated genes and down-regulation of exhaustion-associated genes were similar between research- and large-scale products, suggesting retention of stem-like qualities of LYL845

Figure 9: Research-Scale and Large-Scale LYL845 Productions Demonstrated Comparable Phenotypes

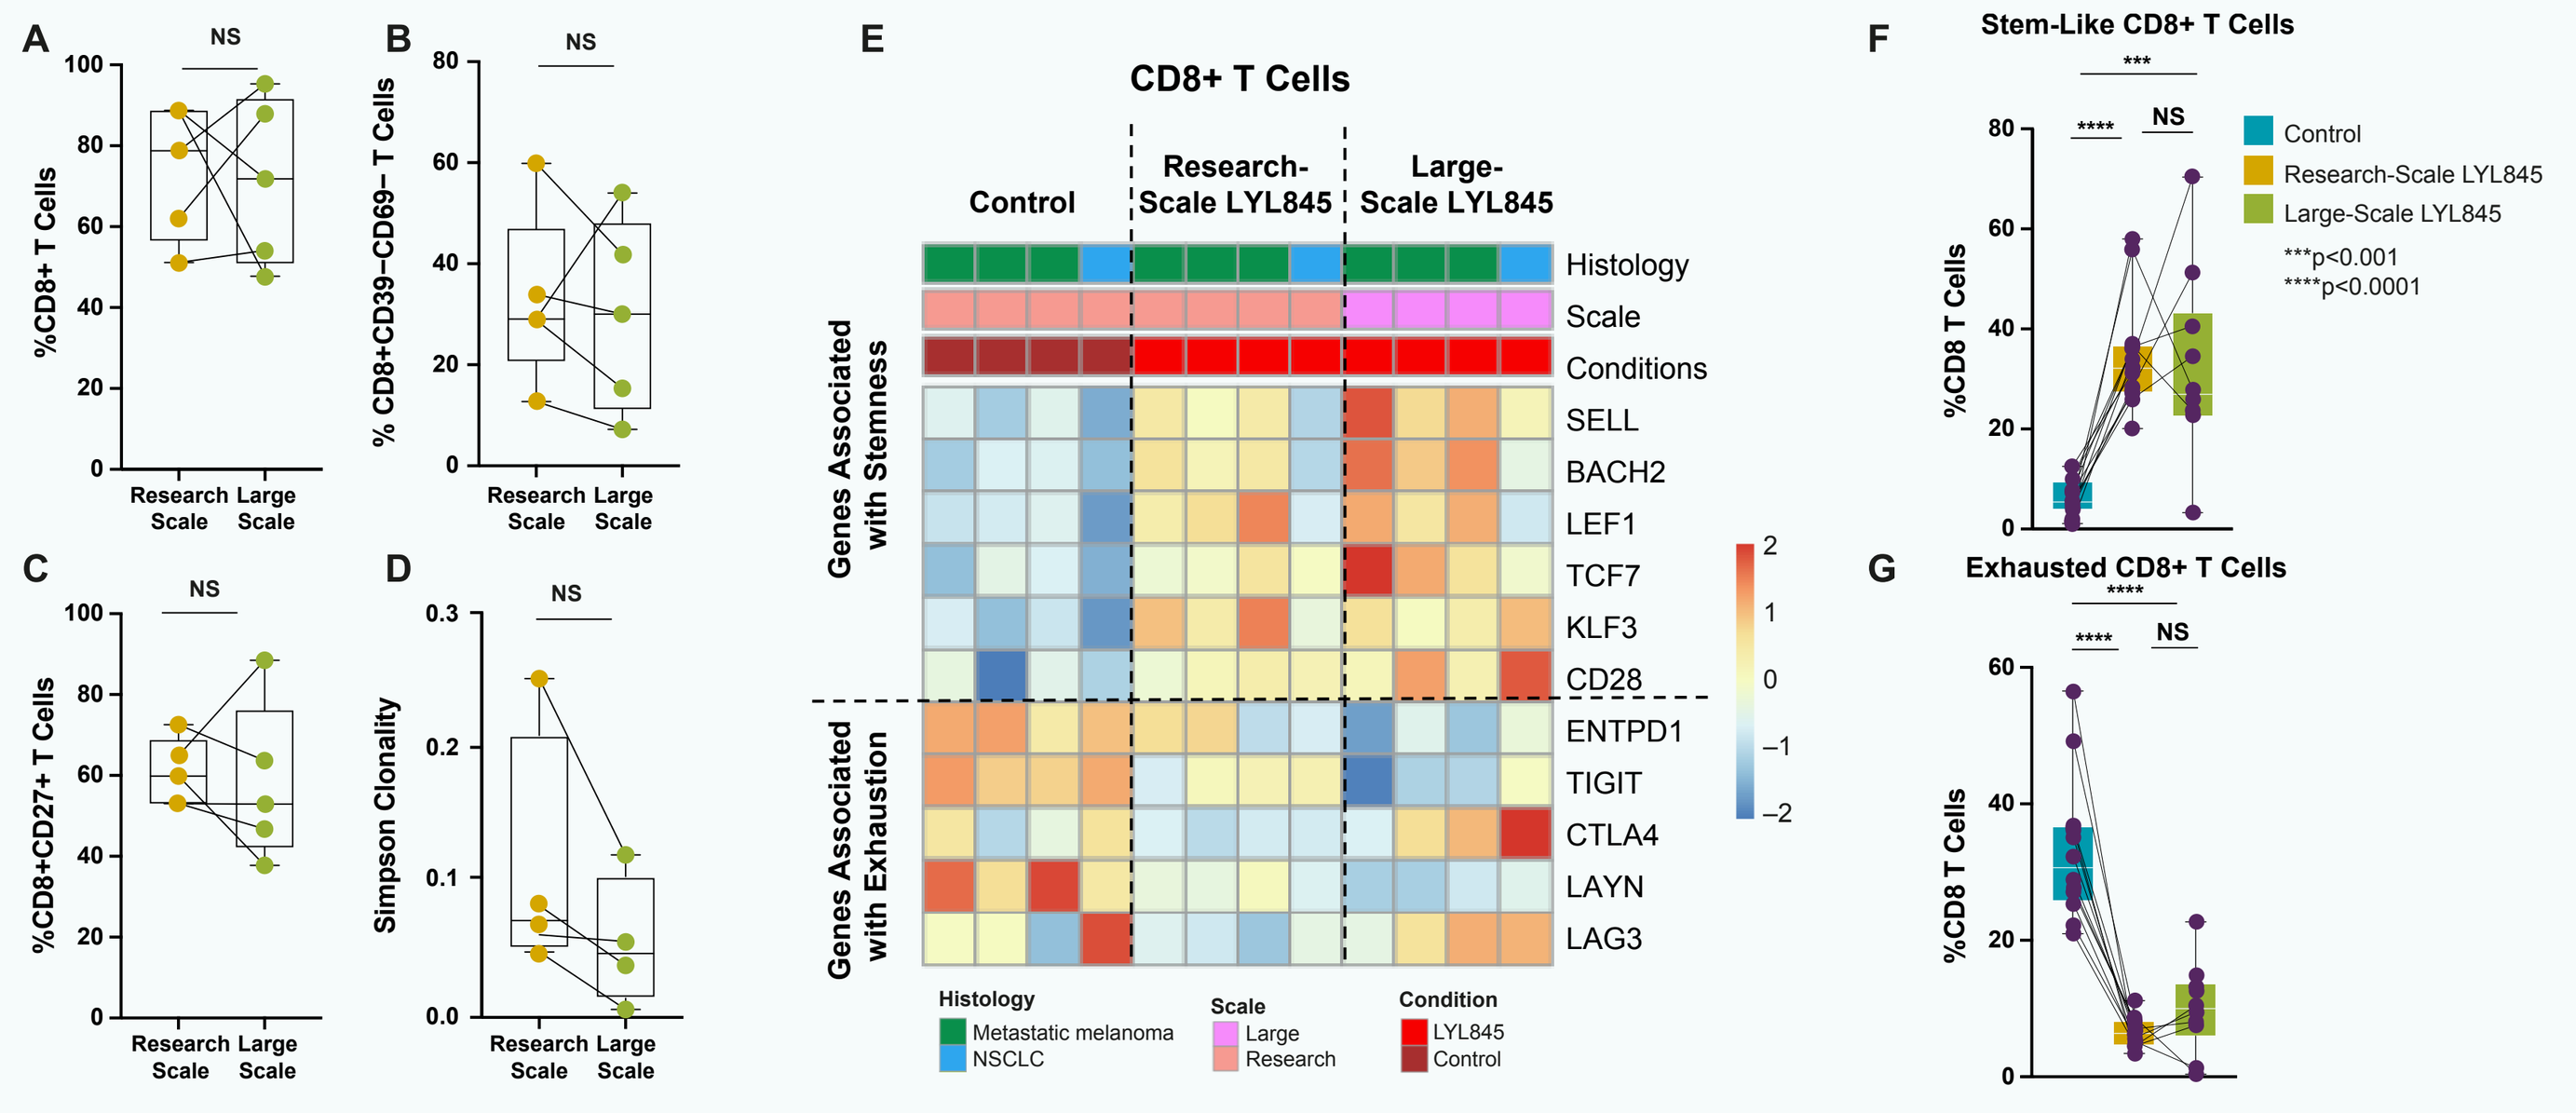
%CD8+ T Cells
% CD8+CD39−CD69− T Cells
%CD8+CD27+ T Cells
Simpson Clonality
Research Scale
Large Scale
CD8+ T Cells
Control
Research-Scale LYL845
Large-Scale LYL845
Genes Associated with Stemness
Genes Associated with Exhaustion
Stem-Like CD8+ T Cells
Exhausted CD8+ T Cells
***p<0.001
****p<0.0001

Target product profiles were compared between both research-scale and large-scale products with similar frequencies in (**A**) CD8+ T cells, (**B**) CD8+CD39−CD69− T cells, (**C**) CD8+CD27+ T cells and (**D**) Simpson Clonality. (**E**) Bulk RNA-seq of research-scale and large-scale Epi-R-produced LYL845 CD8+ T cells have consistently up-regulated stemness-associated genes and down-regulated exhaustion-associated genes when compared with the control processes. Box plot demonstrating (**F**) stem-like and (**G**) exhausted CD8+ T cells identified by single-cell RNA-seq in research-scale control, research-scale LYL845, and large-scale LYL845 products.

Conclusion

- LYL845, produced using the Epi-R protocol, is an investigational autologous TIL product with a greater proportion of CD8+ stem-like T cells, better metabolic fitness, and preserved polyclonality with a higher manufacturing success rate compared with a standard expansion TIL product
- LYL845 maintained stem-like CD8+ T-cell enrichment when manufactured from ICB-refractory melanoma
- Results from LYL845 research- and large-scale productions demonstrate that Epi-R technology enables successful TIL expansion from both immunologically hot and cold tumors
- These data support further development of LYL845, and an Investigational New Drug submission for LYL845 was accepted by the Food and Drug Administration in October 2022

Abbreviations

ACT, adoptive cell therapy; CD, cluster of differentiation; CRC, colorectal cancer; FSCA, forward scatter area; FSCH, forward scatter height; ICB, immune checkpoint blockade; NS, not significant; NSCLC, non-small cell lung cancer; REP, rapid expansion protocol; RNA-seq, RNA sequencing; SSCA, side scatter area; TIL, tumor-infiltrating lymphocyte; TCR, T-cell receptor.

Acknowledgments

The authors would like to thank Lara Zhao, Emily Fu-Sum, and Stefan Siebert for helping with next-generation sequencing and Yi-Dong Lin, Ken Xiong, Andrew Jimena, and Elizabeth Pedroza for flow core services. The authors would like to thank Shannon Christiansen for editorial support. Medical writing and additional editorial support were funded by Lyell Immunopharma and provided by Madison Fagan of BOLDSCIENCE Inc.

References

1. Creelan BC, Wang C, Teer JK, et al. *Nat Med.* 2021;27:1410-1418. **2.** Seitter SJ, Sherry RM, Yang JC, et al. *Clin Cancer Res.* 2021;27:5289-5298. **3.** van den Berg JH, Heemskerk B, van Rooij N, et al. *J Immunother Cancer.* 2020;8:e000848. **4.** Krishna S, Lowery FJ, Copeland AR, et al. *Science.* 2020;370:1328-1334. **5.** Rosenberg SA, Yang JC, Sherry RM, et al. *Clin Cancer Res.* 2011;17:4550-4557. **6.** Jin J, Sabatino M, Somerville R, et al. *J Immunother.* 2012; 35(3):283–292. **7.** Lozano-Rabella M, Gros A. *Clin Cancer Res.* 2020;26:4177–4179. **8.** Vodnala SK, Eil R, Kishton RJ, et al. *Science.* 2019;363:eaau0135.

Presented at SITC Annual Meeting 2022; November 8-12, 2022; Boston, MA