Epi-R™ Technology Produces a Polyclonal TIL Product (LYL845) With Diverse Tumor-Reactive Clones That Have Stem-Like Qualities and Anti-Tumor Function

Abstract 340

Benjamin D. Harris, Yogin Patel, Ngoc-Han Ha, Joanna Kritikou, Lora Zhao, Sheila Lou, Stefan Siebert, Emily Fu-Sum, Rigel Kishton, Purnima Sundar, Suman Kumar Vodnala, Shobha Potluri
Lyell Immunopharma, Inc., South San Francisco, CA and Seattle, WA, USA

Background

- Adoptive cell therapy (ACT) using tumor-infiltrating lymphocytes (TILs) is a promising method for cancer treatment
- TILs that are highly enriched with tumor-reactive T-cell clones have been shown to mediate treatment response in advanced cancers[1-3]
- Conventional TIL products comprise a mixture of extensively differentiated T cells and low levels of stem-like T cells
- Higher proportions of stem-like T cells have been associated with improved outcomes in patients treated with TIL therapies[4]
- LYL845 is an autologous TIL product enhanced with Epi-R™ technology, a proprietary epigenetic reprogramming protocol designed to improve T-cell stemness and anti-tumor functionality in ACT products

Methods

Production of LYL845 Using Epi-R Technology

- LYL845 was prepared with Epi-R technology designed to intentionally and reproducibly generate TILs with durable stemness that can proliferate, persist, and provide prolonged anti-tumor functionality
- The Epi-R protocol is composed of a specifically formulated cell culture media with optimized cytokine composition and cell activation that induces metabolic reprogramming via reduced glycolysis and hypoxia
- LYL845 was produced at research scale using a total of 15 tumor samples (melanoma, lung, and colon) and was compared with control TIL products generated without Epi-R
- LYL845 produced at large scale were generated from nine tumor samples and compared with corresponding research-scale products

Identifying and Tracking Putative Tumor-Reactive Clones (Figure 1)

- Using previously validated methods,[4-5] high-frequency and exhausted clones in the dissociated tumor suspension of Day 0 tumor samples were identified as putative tumor-reactive clones and used to:
 - Identify the top 100 high-frequency TCR clones using bulk TCR-seq
 - Identify exhausted cell TCR clones using single-cell RNA-seq and single-cell TCR-seq
 - Identify a subset of 100 high-frequency and exhausted clones as putative tumor-reactive clones
- Bulk TCR-seq was used to check the presence and proportion of putative tumor-reactive clones in the T-cell products
- Single-cell RNA/TCR-seq was used to evaluate the phenotype of putative tumor-reactive clones in the T-cell products

Experimental Testing of Tumor-Reactive Clones in LYL845 (Figure 1)

- Putative tumor-reactive TCRs were transduced into healthy donor CD8+ T cells and then co-cultured individually against autologous tumor cells. Tumor reactivity was measured by cytolysis and IFNγ secretion 24 hours post co-culture
- TIL products were co-cultured with either patient-dissociated tumor suspension or autologous tumor cells, and tumor reactivity was measured by 4-1BB and IFNγ expression in single-cell RNA/TCR-seq

Figure 1: Methodology for Identifying, Tracking, and Testing Putative Tumor-Reactive Clones

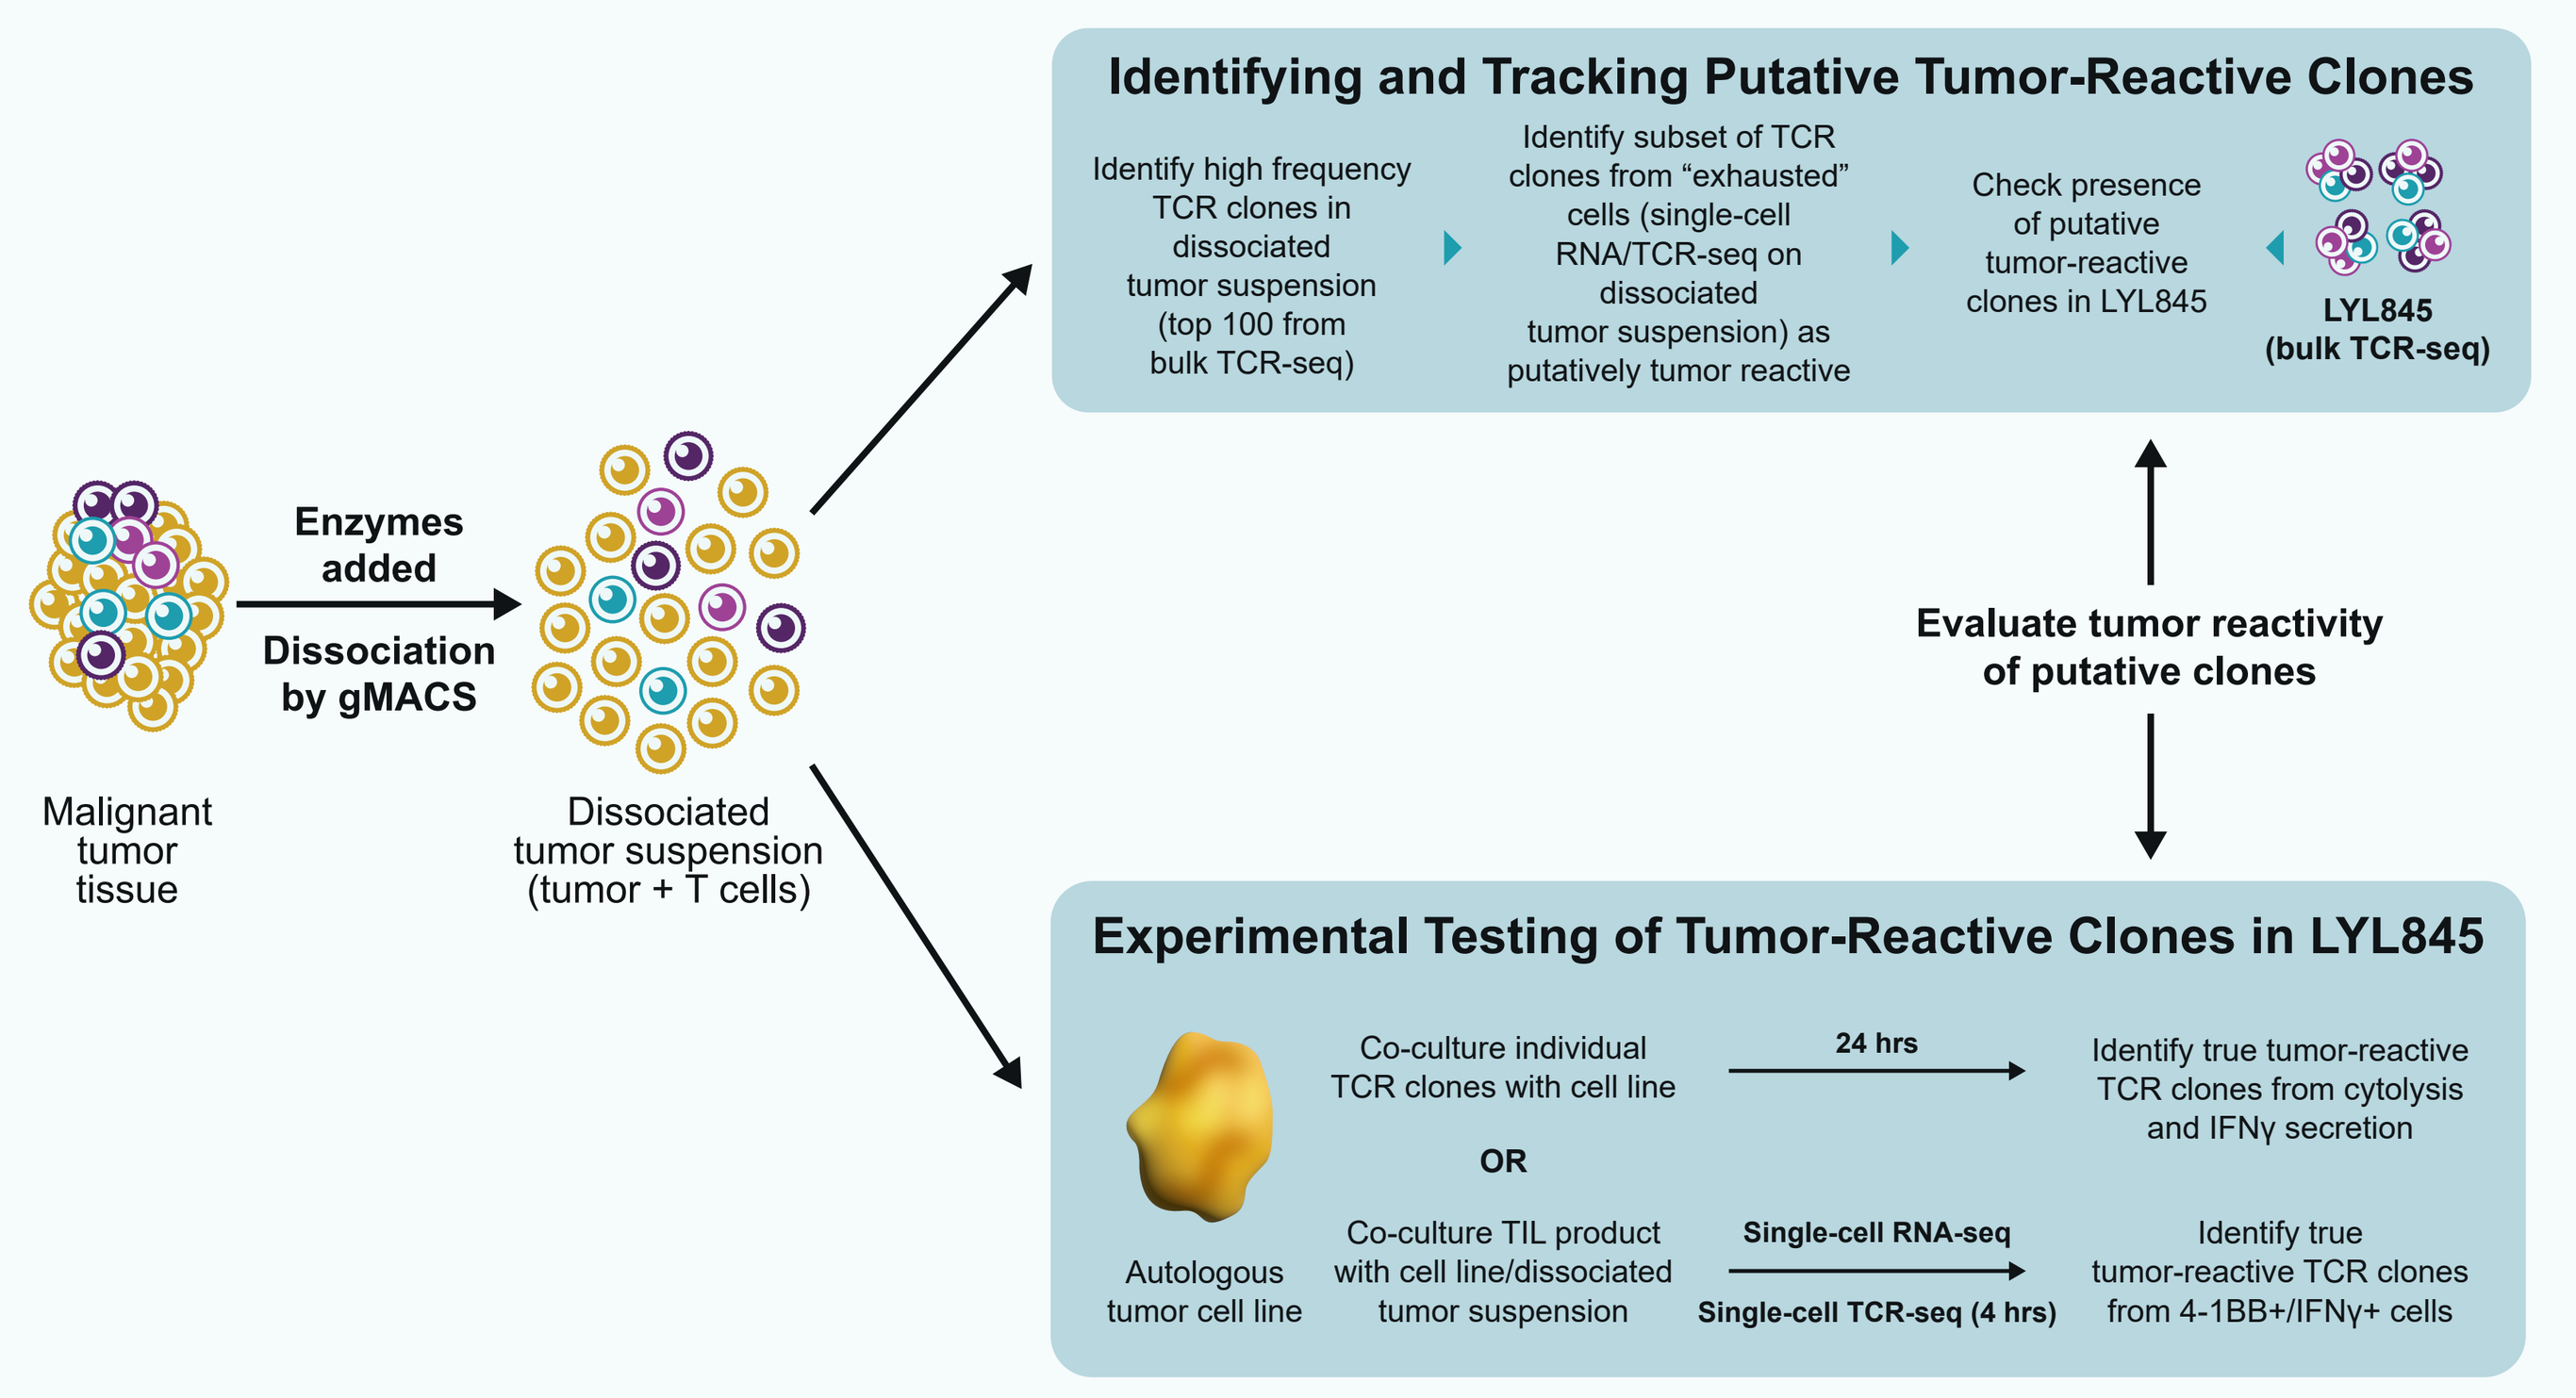
Identifying and Tracking Putative Tumor-Reactive Clones
Identify high frequency TCR clones in dissociated tumor suspension (top 100 from bulk TCR-seq)
Identify subset of TCR clones from "exhausted" cells (single-cell RNA/TCR-seq on dissociated tumor suspension) as putatively tumor reactive
Check presence of putative tumor-reactive clones in LYL845
LYL845 (bulk TCR-seq)
Malignant tumor tissue
Enzymes added
Dissociation by gMACS
Dissociated tumor suspension (tumor + T cells)
Evaluate tumor reactivity of putative clones
Experimental Testing of Tumor-Reactive Clones in LYL845
Autologous tumor cell line
Co-culture individual TCR clones with cell line
24 hrs
Identify true tumor-reactive TCR clones from cytolysis and IFNγ secretion
OR
Co-culture TIL product with cell line/dissociated tumor suspension
Single-cell RNA-seq
Single-cell TCR-seq (4 hrs)
Identify true tumor-reactive TCR clones from 4-1BB+/IFNγ+ cells

Results

Key Findings

Preclinical analysis of the Epi-R-produced TIL product demonstrated that LYL845 exhibited:
- High polyclonality and anti-tumor activity
- Preservation of putative tumor-reactive clones
- Increased stemness and reduced exhaustion-associated genes in putative tumor-reactive clones

LYL845 Products Are Highly Polyclonal and Demonstrate Potent Anti-Tumor Activity

- LYL845 expanded from three tumor types using Epi-R technology demonstrated a high degree of polyclonality (based on the Simpson Clonality Index); LYL845 manufactured at research scale indicated that clonal diversity of source tumor tissue is preserved (**Figure 2A**)
- LYL845 large-scale products demonstrated higher polyclonality compared with research-scale products (**Figure 2B**)
- LYL845 demonstrated potent anti-tumor function through 1) dose-dependent cytolytic activities during co-culture with an autologous tumor cell line, 2) pro-inflammatory cytokine secretion after CD3/CD28 stimulation

Figure 2: LYL845 Retained Polyclonality With Preserved TCR Diversity of Source Tumor Tissue

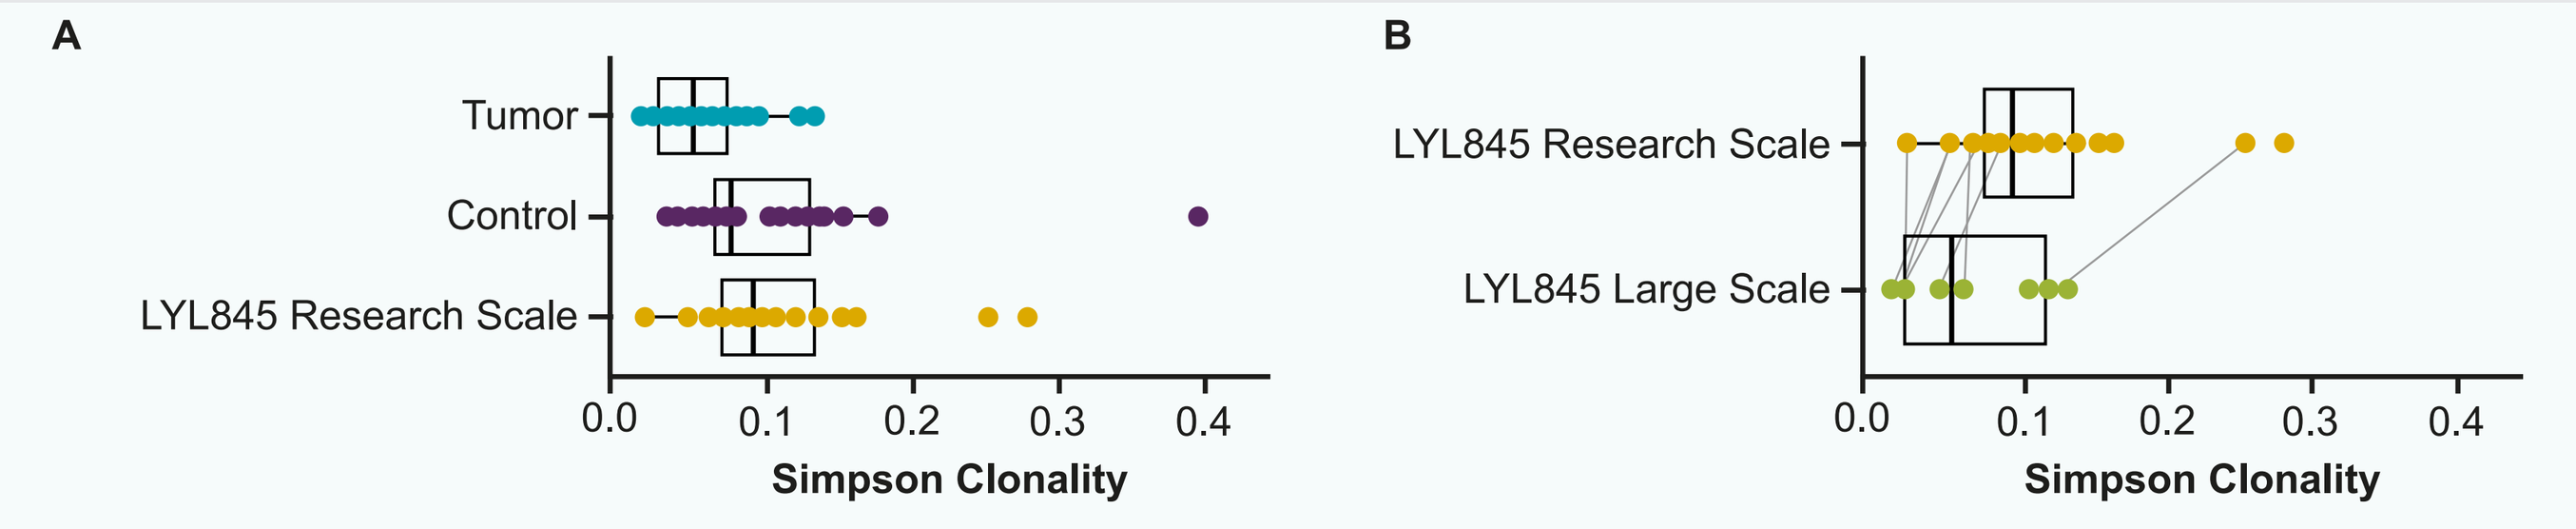
A
Tumor
Control
LYL845 Research Scale
0.0 0.1 0.2 0.3 0.4
Simpson Clonality
B
LYL845 Research Scale
LYL845 Large Scale
0.0 0.1 0.2 0.3 0.4
Simpson Clonality

(**A**) Simpson clonality of dissociated tumor suspension, LYL845 manufactured at research scale, and product from control process at research scale. (**B**) Simpson clonality of large-scale and research-scale LYL845 products.

Figure 3: LYL845 Demonstrated Dose-Dependent Cytolytic Activity and Pro-Inflammatory Cytokine Secretion

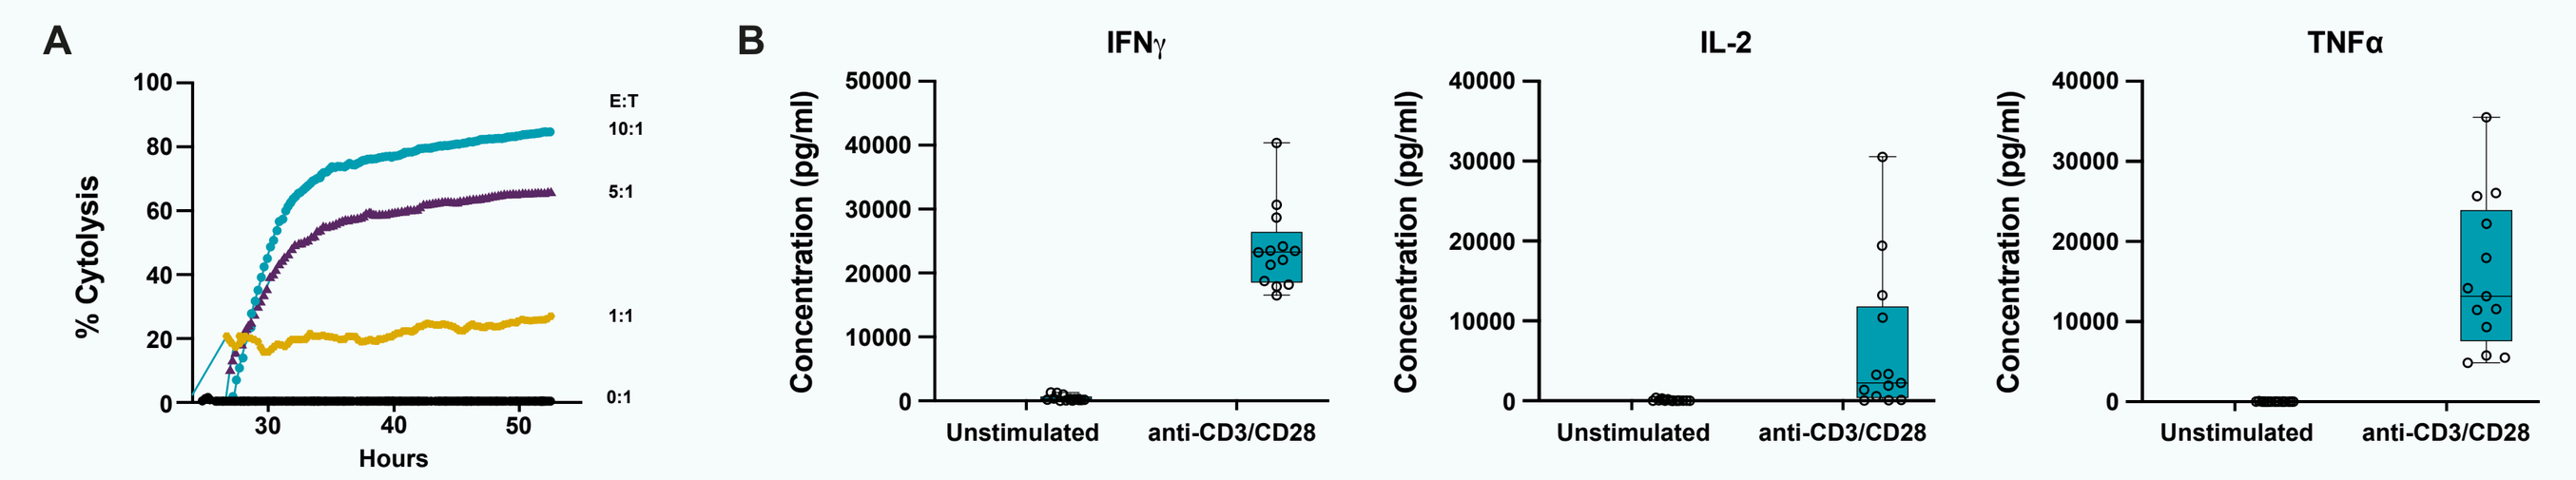
A
% Cytolysis
Hours
E:T
10:1
5:1
1:1
0:1
B
IFNγ
IL-2
TNFα
Concentration (pg/ml)
Unstimulated
anti-CD3/CD28

(**A**) Tumor killing by LYL845 as percent cytolysis with an autologous tumor cell line (metastatic melanoma) with tumor cell killing observed in all E:T ratios compared with control tumor-only (0:1) in a dose-dependent manner. (**B**) Increased IFNγ, IL-2, and TNFα cytokine secretion in LYL845 cells after 24 hours of CD3/CD28 stimulation as compared with unstimulated cells.

Epi-R Created LYL845 Product That Preserved Putative Tumor-Reactive Clones From Initial Tumor Sources

- Putative tumor-reactive clones for each tumor were identified from a dissociated tumor suspension using bulk TCR-seq to identify high-frequency clones and using single-cell RNA/TCR-seq to identify clones with an exhausted phenotype (**Figure 4**)
- Culturing individual putative tumor-reactive TCRs with an autologous tumor cell line confirmed tumor reactivity with cytolysis and IFNγ secretion (**Figure 5**)
- Culturing LYL845 with a dissociated tumor suspension or an autologous cell line validated tumor reactivity of putative tumor-reactive clones (**Figure 6**)
- Using the validated putative tumor reactivity approach, large-scale LYL845 products preserved an average of 94% of putative tumor-reactive clones (**Figure 7A**)
- Both research-scale and large-scale LYL845 products retained a comparable percentage of putative tumor-reactive cells (**Figure 7B**)

Figure 4: Putative Tumor-Reactive Clones Were Identified From Sequencing Thirteen Day-0 Tumors

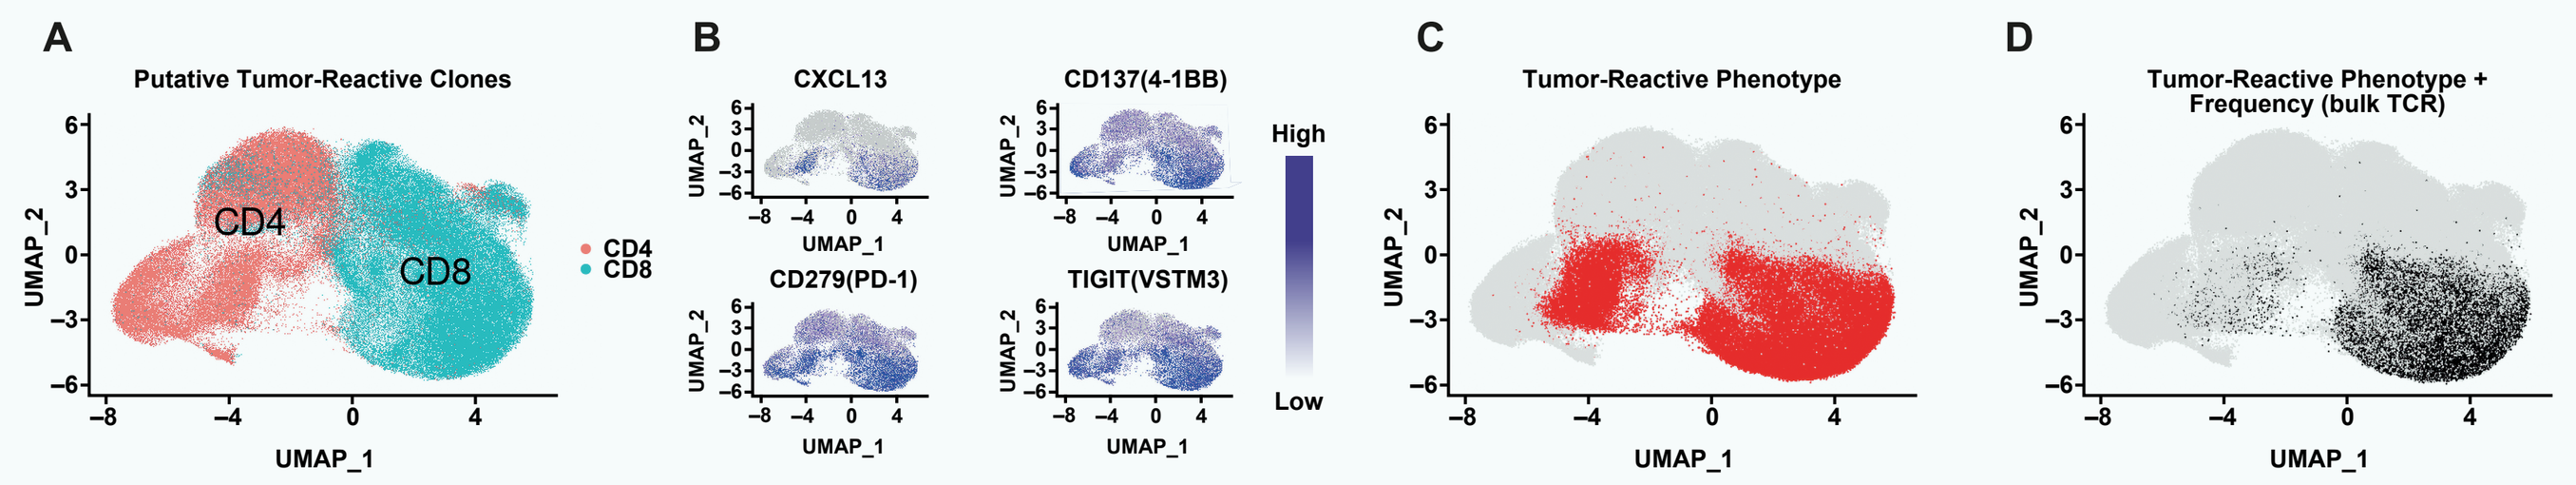
A
Putative Tumor-Reactive Clones
CD4
CD8
UMAP_1
UMAP_2
B
CXCL13
CD137(4-1BB)
CD279(PD-1)
TIGIT(VSTM3)
High
Low
C
Tumor-Reactive Phenotype
D
Tumor-Reactive Phenotype + Frequency (bulk TCR)

(**A**) Analysis by single-cell RNA/TCR-seq of 203,389 cells from 11 melanomas, three lung, and two colon cancers identified putative tumor-reactive clones. (**B, C**) Tumor-reactive phenotypes were identified by CXCL13, 4-1BB, PD-1, and TIGIT expression.[4-7] (**D**) Cells identified as having a tumor-reactive phenotype and high frequency in bulk TCR-seq were defined as being putatively tumor-reactive and biased towards CD8+ T cells.

Figure 5: Testing of Three TCRs With an Autologous Tumor Cell Line Validated Tumor-Reactive Clones

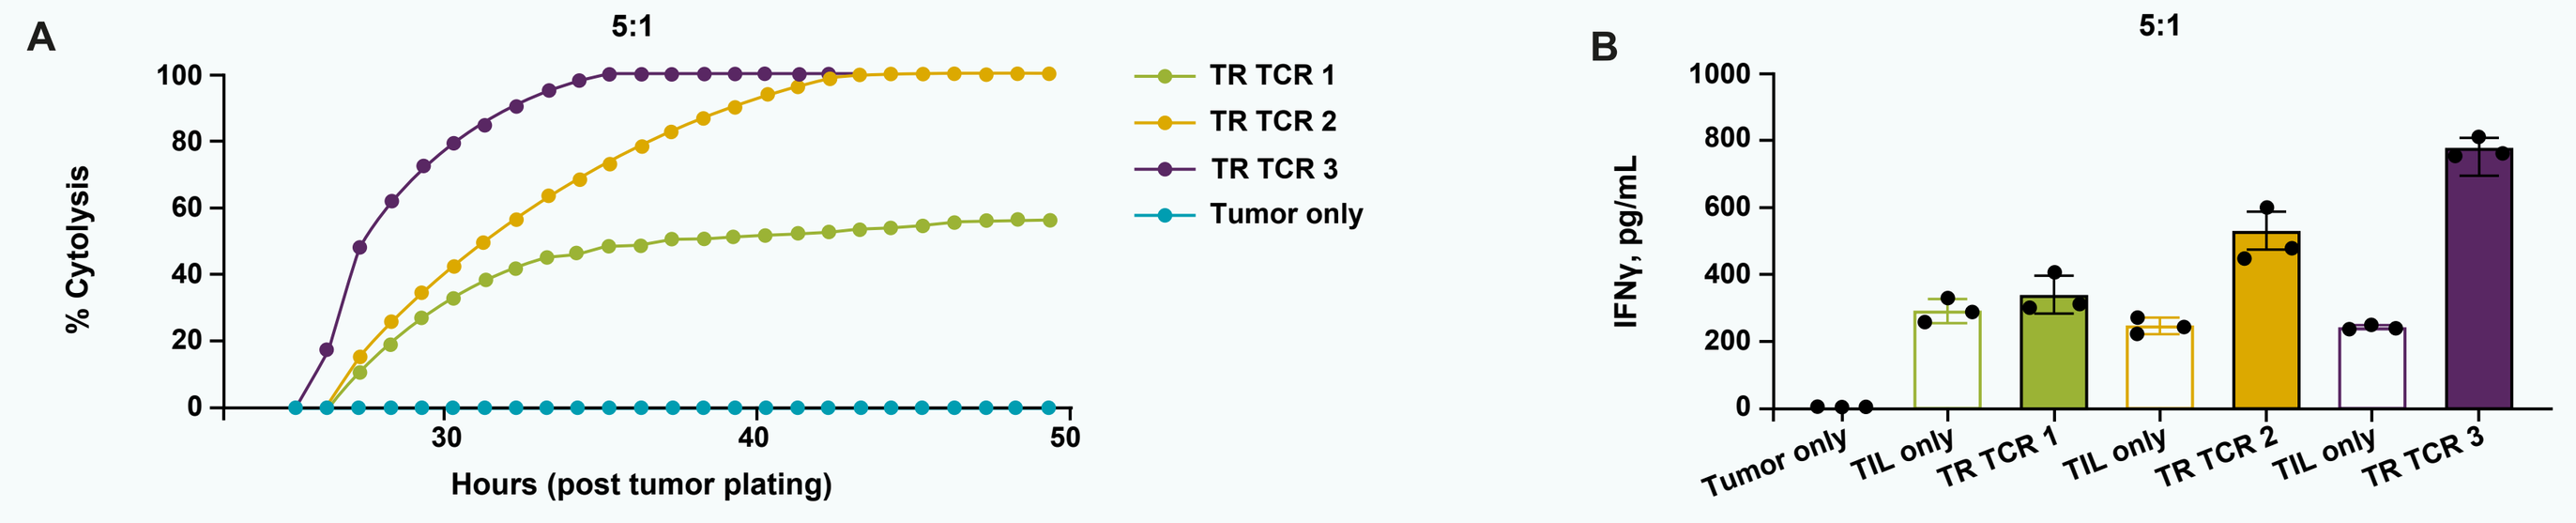
A
5:1
% Cytolysis
Hours (post tumor plating)
TR TCR 1
TR TCR 2
TR TCR 3
Tumor only
B
5:1
IFNγ, pg/mL
Tumor only
TIL only
TR TCR 1
TIL only
TR TCR 2
TIL only
TR TCR 3

The top three putative tumor-reactive TCRs were cloned into healthy donor T cells and co-cultured with the patient's autologous tumor cell line. At an E:T ratio of 5:1, two of the three TCRs showed (**A**) 100% cytolysis which (**B**) correlated with specific IFNγ secretions.

Figure 6: Putative Tumor-Reactive Clones Tested at High Frequencies Were Verified as Tumor Reactive

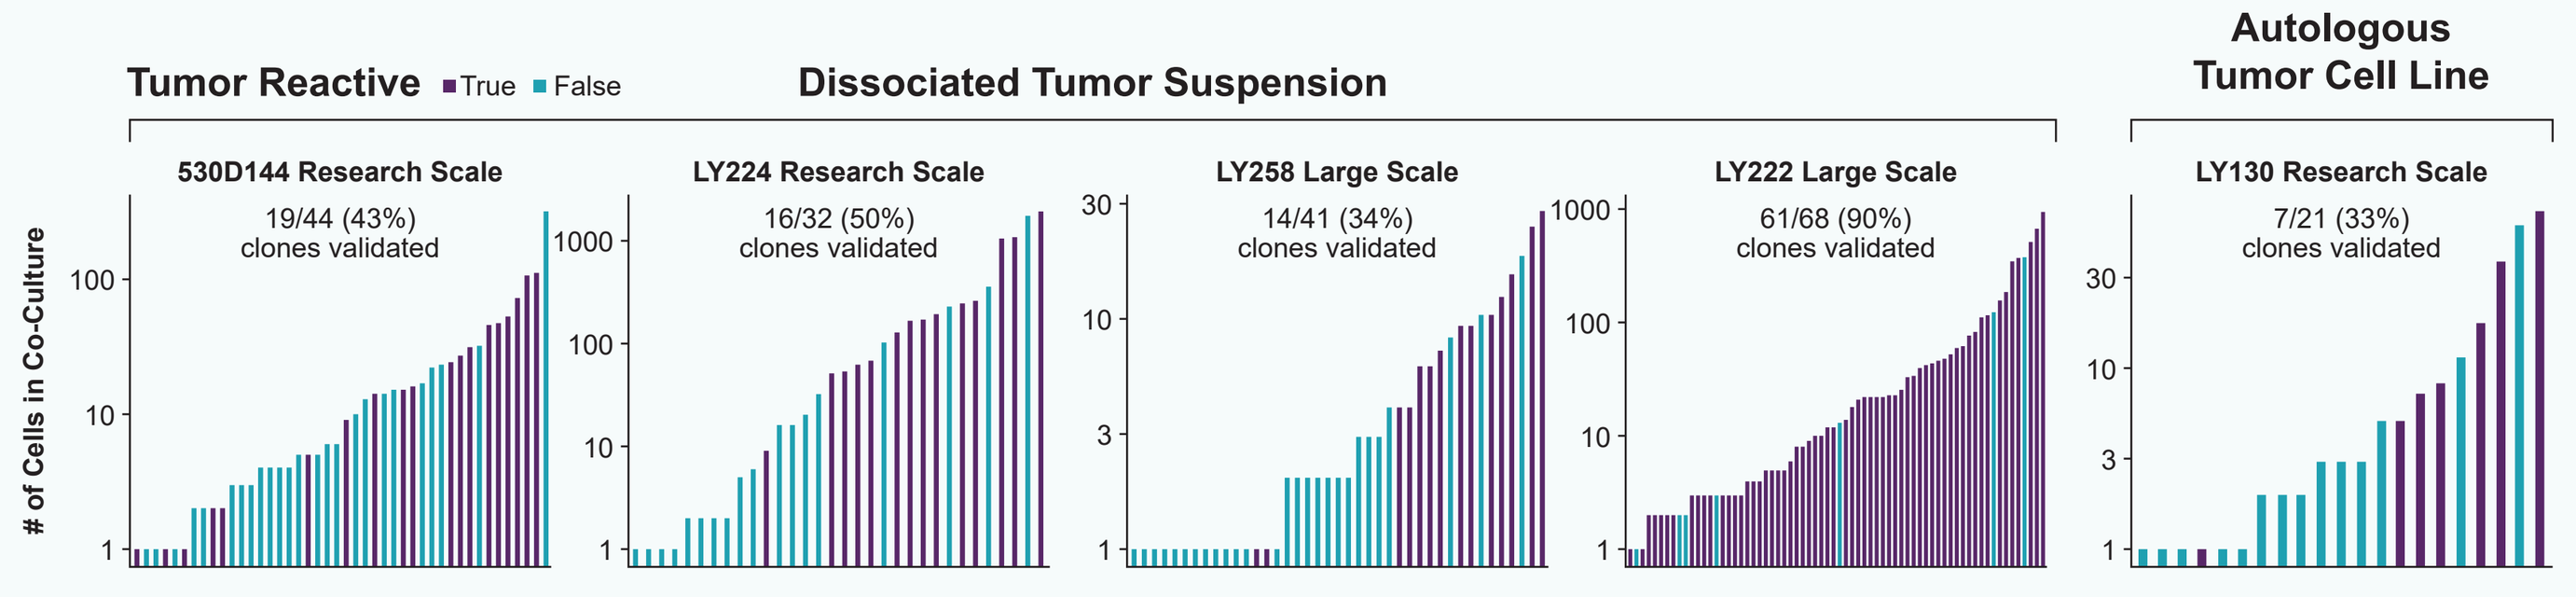
Tumor Reactive
True
False
Dissociated Tumor Suspension
Autologous Tumor Cell Line
530D144 Research Scale
19/44 (43%) clones validated
LY224 Research Scale
16/32 (50%) clones validated
LY258 Large Scale
14/41 (34%) clones validated
LY222 Large Scale
61/68 (90%) clones validated
LY130 Research Scale
7/21 (33%) clones validated
of Cells in Co-Culture

Validation of putative tumor-reactive clones by co-culturing LYL845 with a target autologous tumor cell line or dissociated tumor suspension, then measuring activation (4-1BB+/IFNγ+) showed that putative tumor-reactive clones are tumor-reactive when captured at higher frequencies.

Figure 7: Identified Putative Tumor-Reactive Clones Were Preserved in LYL845 and Control Products

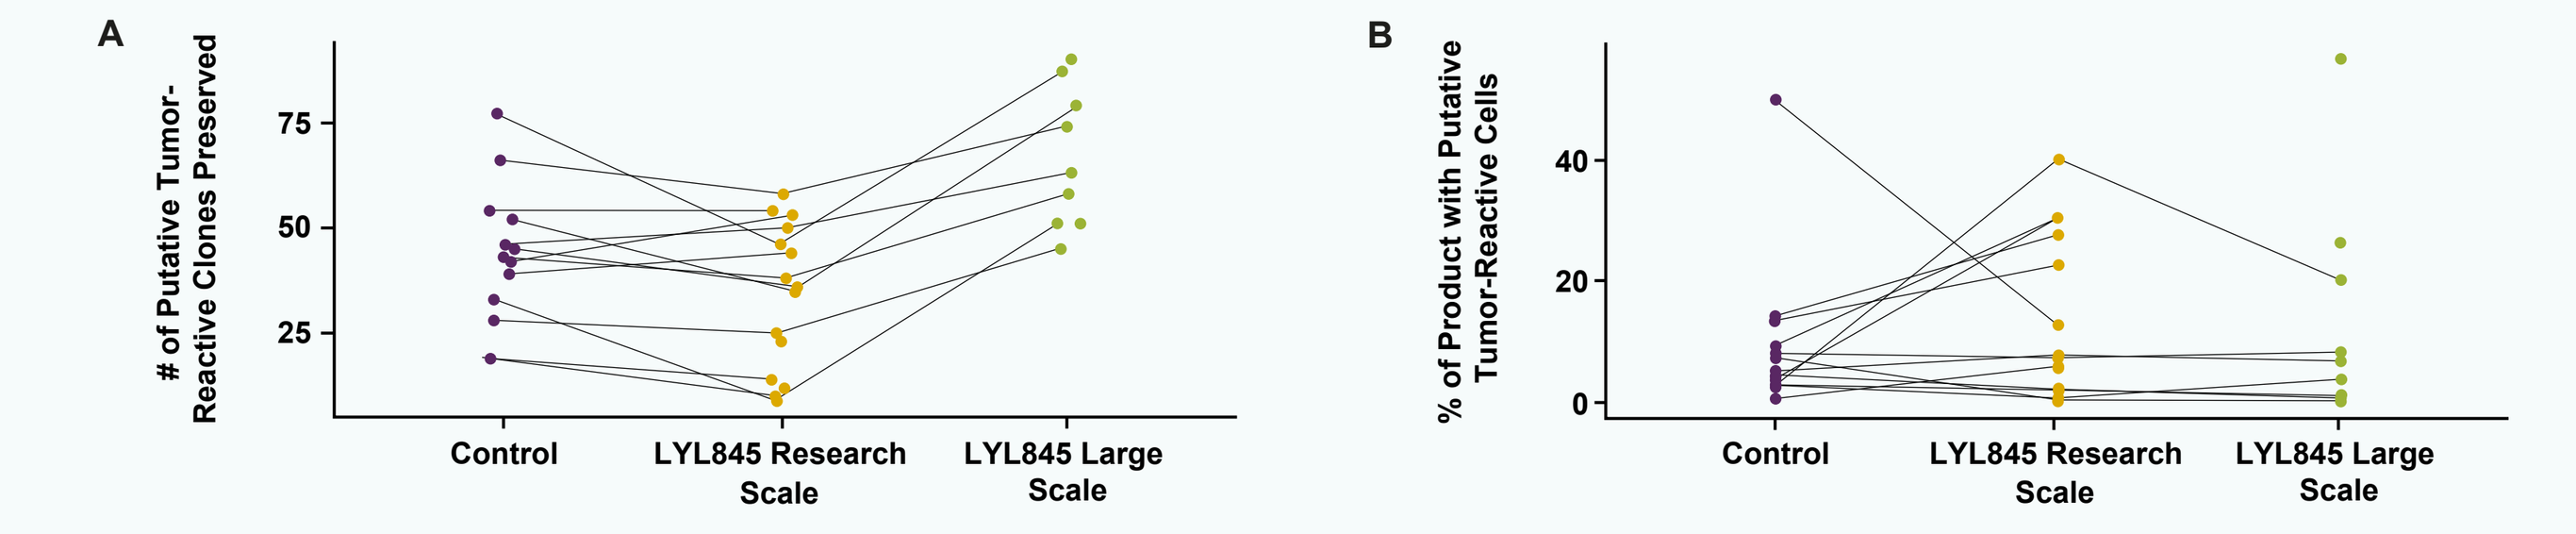
A
of Putative Tumor-Reactive Clones Preserved
Control
LYL845 Research Scale
LYL845 Large Scale
B
% of Product with Putative Tumor-Reactive Cells

Bulk TCR-seq of research-scale products showed that LYL845 and T cells prepared with the control protocol preserved a comparable number of putative tumor-reactive clones. (**A**) Large-scale production of LYL845 preserved 94% of the identified putative tumor-reactive clones versus 57% in research-scale products. (**B**) Both research-scale and large-scale preparations of LYL845 retained a large percentage of putative tumor-reactive cells.

Putative Tumor-Reactive Clones in LYL845 Demonstrated Increased Stem-Like Characteristics With Reduced Expression of Exhaustion Related Genes

- Single-cell RNA/TCR-seq of LYL845 and control TILs demonstrated enrichment of putative tumor-reactive clones in non-stem-like compartments (**Figure 8**)
- Putative tumor-reactive clones in LYL845 demonstrated up-regulation of genes associated with stemness and down-regulation of genes associated with exhaustion when compared with TIL products manufactured using the control process, a trend also seen in large-scale LYL845 production (**Figure 9**)

Figure 8: Putative Tumor-Reactive Cells Were Largely Enriched in Non-Stem-Like Compartments

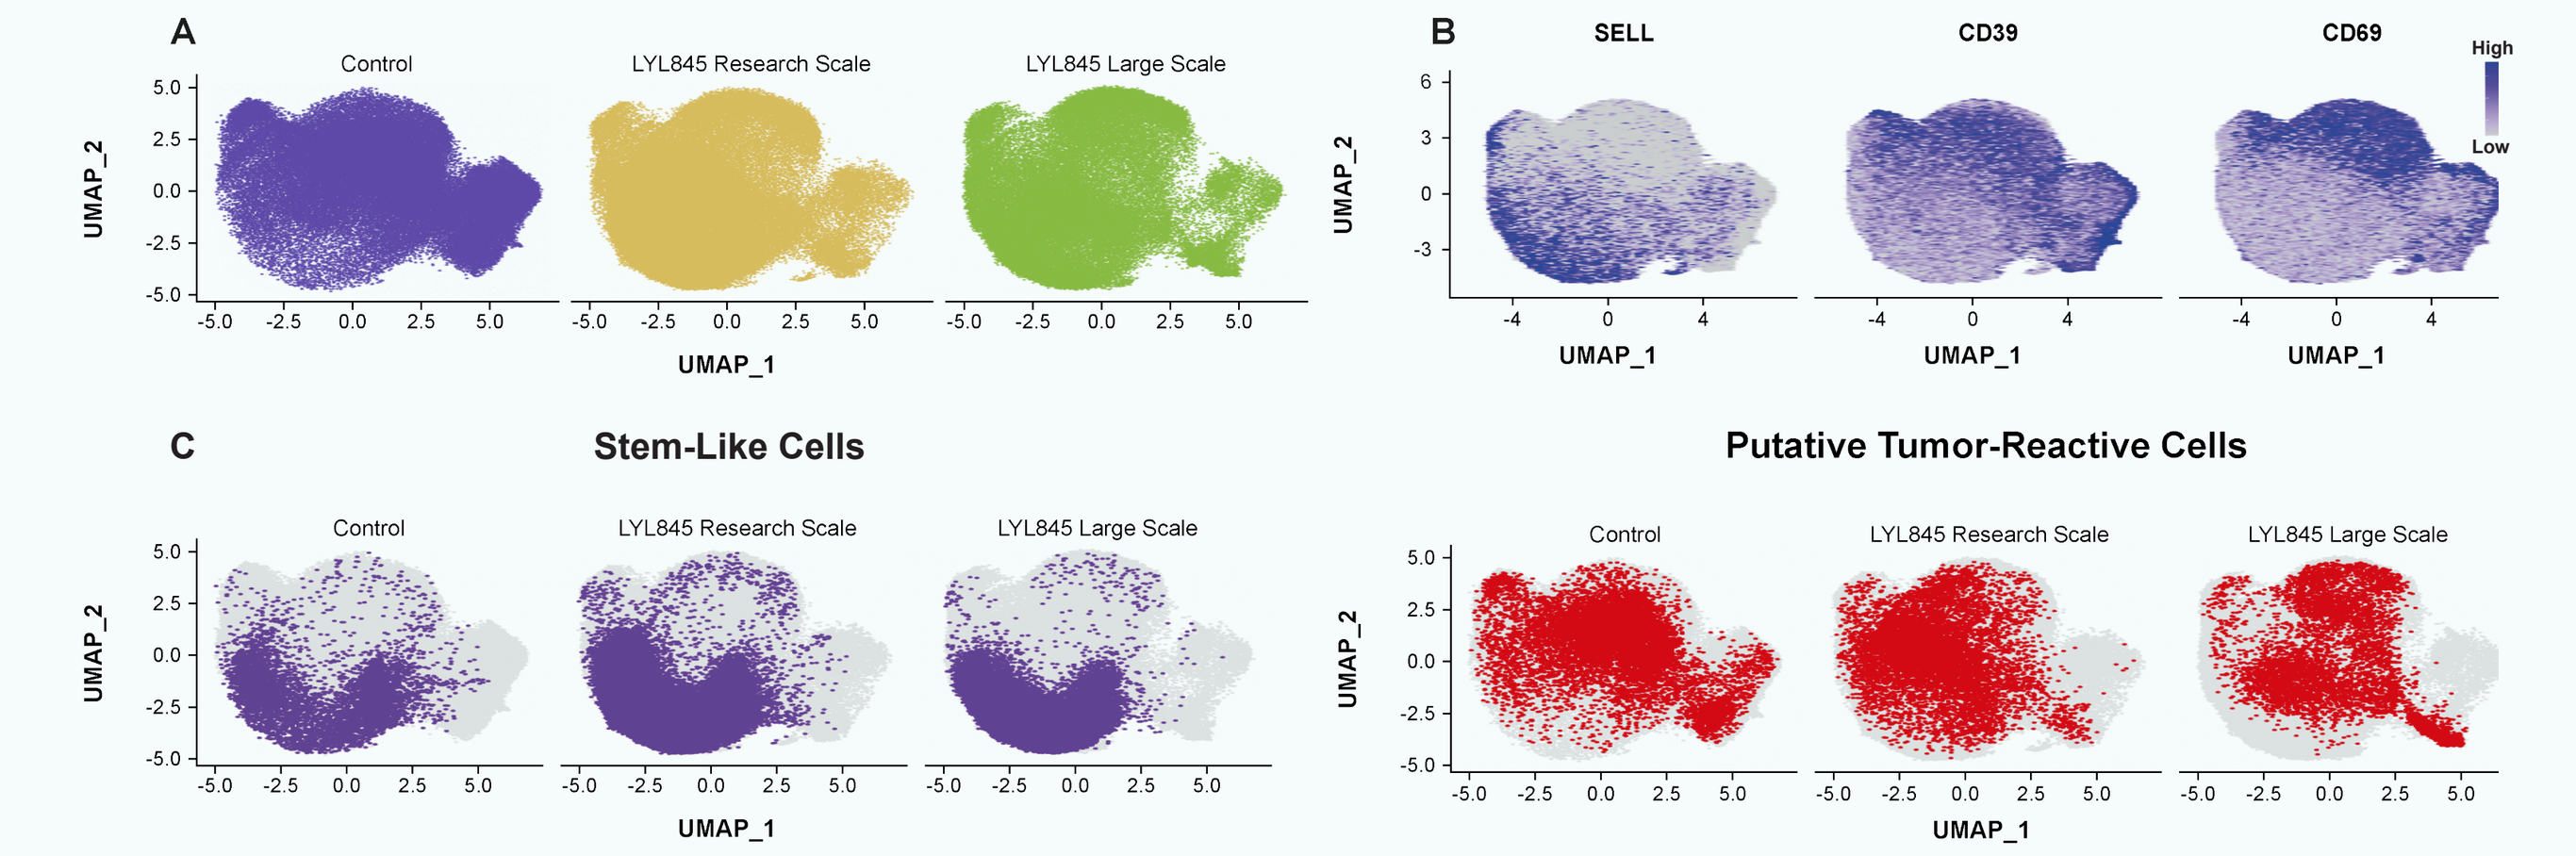
A
Control
LYL845 Research Scale
LYL845 Large Scale
UMAP_1
UMAP_2
B
SELL
CD39
CD69
High
Low
C
Stem-Like Cells
Putative Tumor-Reactive Cells

(**A**) Analysis by single-cell RNA-seq/single-cell TCR-seq of 25 products (nine control, ten LYL845 research-scale, and six LYL845 large-scale) enabled phenotypic characterization of putative tumor-reactive clones. (**B**) Stem-like clusters were identified by expression of SELL, CD39, CD69, and expression of stemness gene sets from literature.[8] (**C**) Putative tumor-reactive clones were enriched in non-stem-like clusters.

Figure 9: LYL845 Putative Tumor-Reactive Cells Had Up-Regulation of Stemness Genes and Down-Regulation of Exhaustion Genes Compared With Control TIL

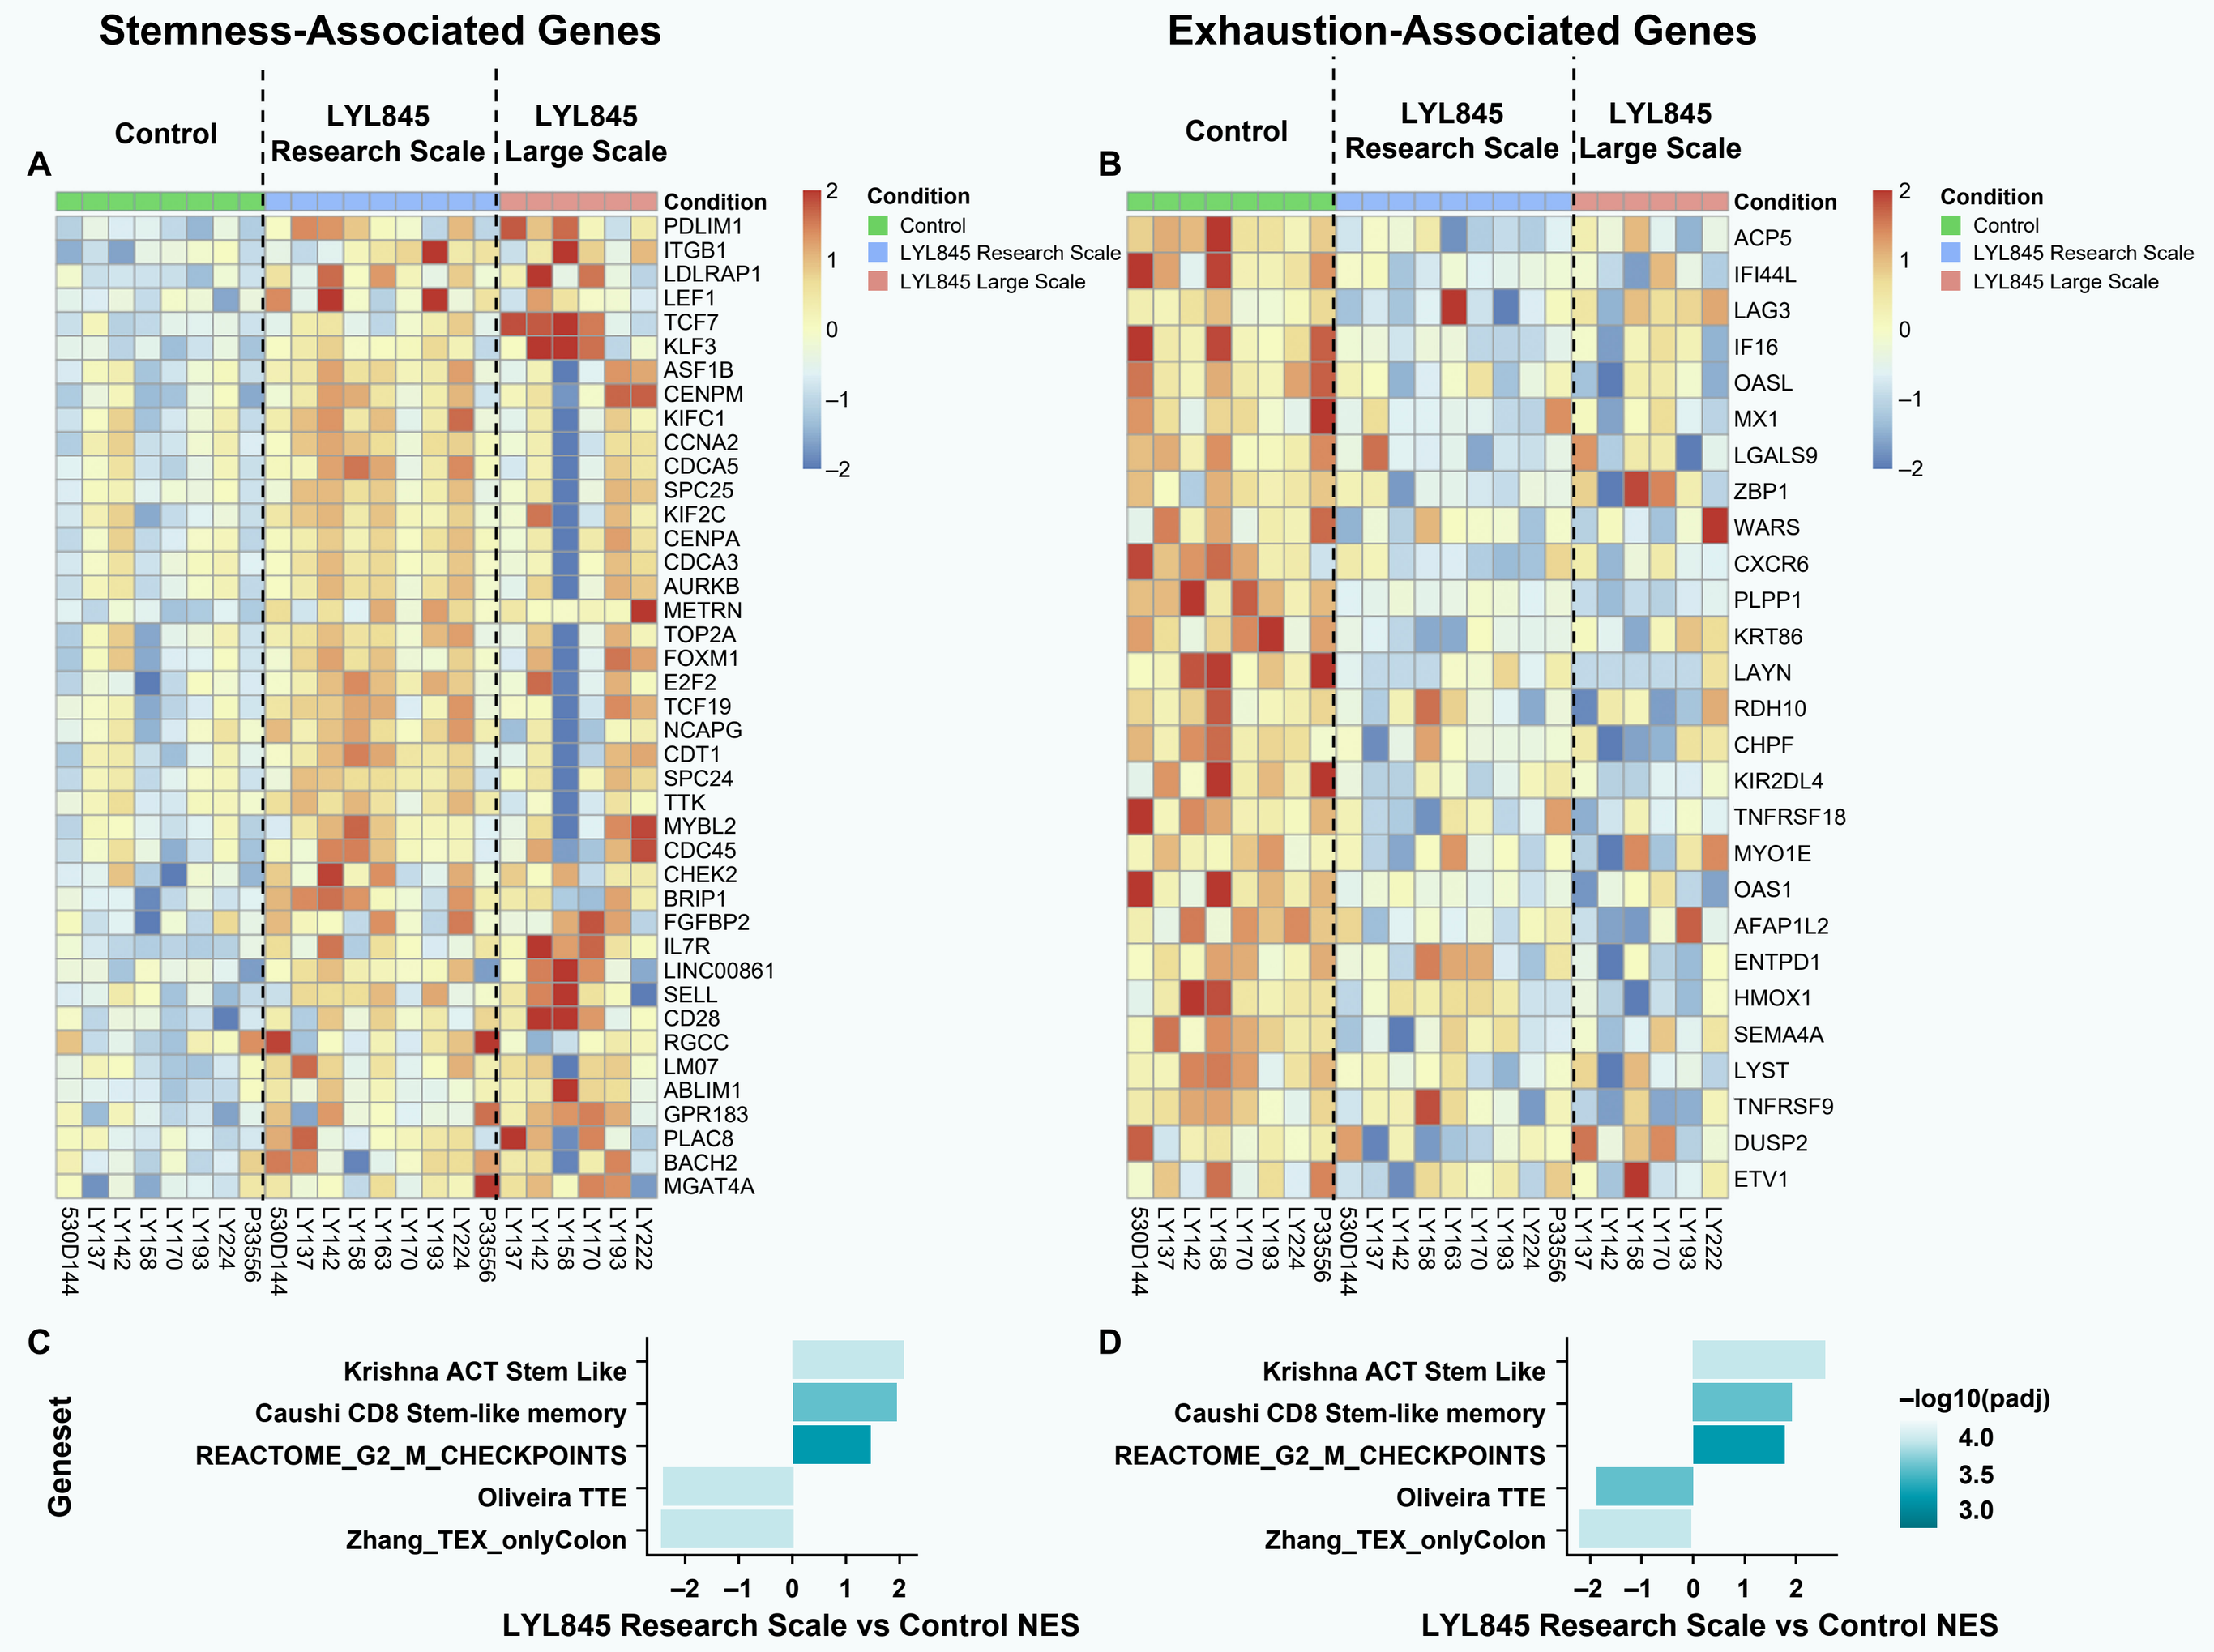
Stemness-Associated Genes
Exhaustion-Associated Genes
A
B
Control
LYL845 Research Scale
LYL845 Large Scale
Condition
PDLIM1
ITGB1
LDLRAP1
LEF1
TCF7
KLF3
ASF1B
CENPM
KIFC1
CCNA2
CDCA5
SPC25
KIF2C
CENPA
CDCA3
AURKB
METRN
TOP2A
FOXM1
E2F2
TCF19
NCAPG
CDT1
SPC24
TTK
MYBL2
CDC45
CHEK2
BRIP1
FGFBP2
IL7R
LINC00861
SELL
CD28
RGCC
LM07
ABLIM1
GPR183
PLAC8
BACH2
MGAT4A
ACP5
IFI44L
LAG3
IF16
OASL
MX1
LGALS9
ZBP1
WARS
CXCR6
PLPP1
KRT86
LAYN
RDH10
CHPF
KIR2DL4
TNFRSF18
MYO1E
OAS1
AFAP1L2
ENTPD1
HMOX1
SEMA4A
LYST
TNFRSF9
DUSP2
ETV1
C
D
Geneset
Krishna ACT Stem Like
Caushi CD8 Stem-like memory
REACTOME_G2_M_CHECKPOINTS
Oliveira TTE
Zhang_TEX_onlyColon
LYL845 Research Scale vs Control NES
−log10(padj)
4.0
3.5
3.0

(**A, B**) Pseudobulk differential expression between putative tumor-reactive cells in LYL845 research-scale and control products indicated up-regulation of stemness- and down-regulation of exhaustion-associated genes and gene sets from literature. The large-scale preparation of LYL845 putative tumor-reactive cells had a comparable phenotype to the research-scale preparation. (**C, D**) GSEA identified up-regulation of stemness-associated gene sets from literature and down-regulation of exhaustion-associated genes in both research- and large-scale preparations of LYL845 compared with control.[4-5,9-10]

Conclusion

- Preclinical analysis of LYL845 revealed a highly polyclonal product with potent anti-tumor activity
- LYL845 demonstrated dose-dependent cytolytic activity and cytokine secretion when co-cultured with an autologous melanoma cell line
- Preclinical data showed that LYL845 has preserved tumor-reactive clones with stem-like qualities
- These promising preclinical data led to the further development of LYL845, an investigational autologous TIL therapy enhanced with Epi-R
- An Investigational New Drug application for LYL845 was accepted by the FDA in October 2022, and it will be evaluated for safety, tolerability, and anti-tumor activity in an upcoming Phase 1 clinical trial

Abbreviations

4-1BB, tumor necrosis factor ligand superfamily member 9; ACT, adoptive cell therapy; CD, cluster of differentiation; CXCL, chemokine ligand; E:T, effector:target; gMACS, gentle magnetic-activated cell sorting; GSEA, gene set enrichment analysis; IFNγ, interferon gamma; IL-2, interleukin-2; NES, normalized enrichment score; PD-1, programmed cell death protein 1; RNA, ribonucleic acid; RNA-seq, ribonucleic acid sequencing; TCR, T-cell receptor; TCR-seq, T-cell receptor sequencing; TIGIT, T-cell immune receptor with immunoglobin and ITIM domains; TIL, tumor-infiltrating lymphocyte; TNFα, tumor necrosis factor alpha; TR, tumor-reactive; UMAP, uniform manifold approximation and projection.

Acknowledgments

We thank members of Lyell Immunopharma's flow cytometry core (Andrew Jimena, Elizabeth Pedrosa, Ken Xiong), process development team (Meri Galindo Casas, Carson Harms, Melissa DeFrancesco), and Queenie Vong for their experimental contributions. Medical writing and editorial support were funded by Lyell Immunopharma and provided by Madison Fagan of BOLDSCIENCE Inc.

References

1. van den Berg JH, Heemskerk B, van Rooij N, et al. *J Immunother Cancer*. 2020;8:e000848. **2.** Tran E, Turcotte S, Gros A, et al. *Science*. 2014;344:641-645. **3.** Zacharakis N, Chinnasamy H, Black M, et al. *Nat Med*. 2018;24:724-730. **4.** Krishna S, Lowery FJ, Copeland AR. *Science*. 2020;370:1328-1334. **5.** Oliveira G, Stromhaug K, Klaeger S, et al. *Nature*. 2021;596:119-125. **6.** Pasetto A, Gros A, Robbins PF, et al. *Cancer Immunol Res*. 2016;4:734-743. **7.** Lowery FJ, Krishna S, Yossef R, et al. *Science*. 2022; 875:877-884. **8.** Caushi JX, Zhang J, Ji Z, et al. *Nature*. 2021;596:126-32. Erratum in: *Nature*. 2021;598:E1. **9.** Zhang L, Yu X, Zheng L. *Nature*. 2018;564:268-272. **10.** Sade-Feldman M, Yizhak K, Bjorgaard SL, et al. *Cell*. 2018;175:998-1013.

Presented at SITC Annual Meeting 2022; November 8-12, 2022; Boston, MA